ASSOCIATION BETWEEN CENTRAL PRECOCIOUS PUBERTY AND COMORBID MEDICAL ILLNESS: LARGE-SCALE RETROSPECTIVE CLAIMS ANALYSIS OF FLORIDA MEDICAID-ENROLLED CHILDREN

Hankin C[1], Silverman L[2], Wang Z[1], Bronstone A[1]

(1) BioMedEcon, LLC, South San Francisco, CA, USA; (2) Goryeb Children's Hospital, Morristown, NJ, USA

OBJECTIVES

Central precocious puberty (CPP) is a rare disorder in children characterized by premature reactivation of the hypothalamus-pituitary-gonadal (HPG) axis, causing early puberty and the development of secondary sexual characteristics, accelerated growth, and accelerated bone maturation.[1]

A small body of research links CPP with increased risk for central nervous system and congenital disorders,[2] but little is known about the association of CPP with other medical conditions.

We explored a large-scale, retrospective claims database (Florida Medicaid) to examine potential associations of precocious puberty with other medical conditions.

METHODS

Data were obtained from Florida Medicaid computerized claims records, which contain basic demographic information; ICD and CPT diagnosis and treatments codes; and payment data. Information is patient de-identified and fully compliant with the HIPPA Privacy Rule. Subjects were identified from enrollees in the Florida Medicaid program who had a paid claim from July 1997 through June 2004.

We compared children aged 3-7 years with precocious puberty (ICD-9 259.1, which may include premature adrenarche in addition to CPP) to those aged 3-7 years without this diagnosis. Patients were age-, sex-, race-, and year-matched to controls in the year of precocious puberty diagnosis (one-to-many).

ICD-9 diagnoses were used to classify disease groups and disease burden (Charlson Index).[3] The Charlson index is a weighted index of comorbidity that is widely used in retrospective claims analysis.[4] Fisher's exact test was used to examine differences between groups and logistic regression to examine likelihood estimates.

RESULTS

- Among 720,931 children, 1,644 (0.23%) were diagnosed with precocious puberty.
- There was a significant difference in disease burden between groups (p<0.0001).
- After adjustments, compared to Medicaid-enrolled children without the disorder, those with precocious puberty were:
 - 6 times more likely to be diagnosed with hemiplegia (OR 6.3, 95% CI 3.8-10.4, p<0.0001);
 - 5 times more likely to be diagnosed with cerebrovascular disease (OR 5.1, 95% CI 3.2-8.1, p<0.0001) or moderate or severe renal disease (OR 4.7, 95% CI 2.2-9.9, p<0.0001);
 - 4 times more likely to be diagnosed with congestive heart failure (OR 3.9, 95% CI 2.1-7.0, p<0.0001), connective tissue disease (OR 3.7, 95% CI 1.4-9.8, p=0.009), or diabetes (OR 3.5, 95% CI 2.0-6.2, p<0.0001); and
 - 3 times more likely to be diagnosed with chronic pulmonary disease (OR 3.1, 95% CI 2.2-4.3, p<0.0001) or AIDS (OR 2.8, 95% CI 1.6-4.9, p=0.0002).

	CHARLSON WEIGHT	CPP PATIENTS N (%) n=1,644	OTHER PATIENTS N (%) n=7,558,497	P FISHER'S EXACT TEST	LOGISTIC REGRESSION (CONDITION YES VS NO)	
					OR (95% CI)	P-VALUE
MYOCARDIAL INFARCT	1	1 (0.06%)	708 (0.01%)	0.1429		
CONGESTIVE HEART FAILURE	1	11 (0.7%)	9004 (0.1%)	<0.0001	3.869 (2.137 -7.005)	<0.0001
PERIPHERAL VASCULAR DISEASE	1	0	580 (0.01%)	1.0000		
CEREBROVASCULAR DISEASE	1	18 (1.1%)	11169 (0.2%)	<0.0001	5.073 (3.177-8.099)	<0.0001
DEMENTIA	1	1 (0.1%)	879 (0.01%)	0.1742		
CHRONIC PULMONARY DISEASE	1	35 (2.1%)	46154 (0.6%)	<0.0001	3.050 (2.183-4.263)	<0.0001
CONNECTIVE TISSUE DISEASE	1	4 (0.2%)	4117 (0.1%)	0.0132	3.702 (1.392-9.849)	0.0087
ULCER DISEASE	1	1 (0.06%)	810 (0.01%)	0.1617		
MILD LIVER DISEASE	1	1 (0.06%)	772 (0.01%)	0.1547		
DIABETES	1	12 (0.7%)	14188 (0.2%)	<0.0001	3.525 (1.997-6.223)	<0.0001
HEMIPLEGIA	2	16 (1.0%)	4762 (0.1%)	<0.0001	6.306 (3.831-10.382)	<0.0001
DIABETES WITH END-ORGAN DAMAGE	2	1 (0.06%)	729 (0.01%)	0.1468		
MODERATE OR SEVERE RENAL DISEASE	2	7 (0.4%)	4762 (0.1%)	<0.0001	4.688 (2.227-9.869)	<0.0001
ANY TUMOR	2	1 (0.06%)	913 (0.01%)	0.1803		
LEUKEMIA	2	0	794 (0.01%)	1.0000		
LYMPHOMA	2	0	755 (0.01%)	1.0000		
MODERATE-SEVERE LIVER DISEASE	3	0	541 (0.01%)	1.0000		
METASTATIC SOLID TUMOR	6	1 (0.06%)	1817 (0.02%)	0.3266		
AIDS	6	13 (0.8%)	15830 (0.2%)	<0.0001	2.839 (1.643-4.907)	0.0002

CONCLUSIONS

This is the first large-scale analysis to show a strong association between ICD-9 claims-based diagnoses of precocious puberty (which may include premature adrenarche in addition to CPP) and medical disorders. Although researchers have examined the psychosocial consequences of precocious puberty, this study found that precocious puberty significantly increased the likelihood of children developing comorbid medical conditions, including hemiplegia, renal disease, congestive heart failure, cardiopulmonary disease, connective tissue disorders, diabetes, and AIDS. The mechanisms by which children with this hormonal disorder become at increased risk for these particular medical conditions should be explored.

REFERENCES

1. Antoniazzi F, Zamboni G. Central precocious puberty: current treatment options Pediatr Drugs 2004; 6:211-31.
2. Nebesio TD, Eugster EA. Current concepts in normal and abnormal puberty. Curr Probl Pediatr Adolesc Health Care 2007;37:50-72.
3. Charlson ME, Pompei P, Ales KL, MacKenzie CR. A new method of classifying prognostic comorbidity in longitudinal studies: Development and validation. J Chron Dis 1987;40:373-383.
4. Farley JF, Harley CR, Devine JW. A comparison of comorbidity measurements to predict healthcare expenditures. Am J Manag Care 2006;12:110-9.

Research funded by
Indevus Pharmaceuticals, Inc.

BioMedEcon
Health Economics & Outcomes Research

For more information, please contact:
Cheryl Hankin, PhD
President and Chief Scientific Officer, BioMedEcon, LLC
PO Box 129, Moss Beach, CA 94038
cherylhankin@biomedecon.com; 650 563 9475

PRESENTED AT THE 10th ANNUAL EUROPEAN CONGRESS OF ISPOR; DUBLIN, IRELAND; OCTOBER 20-23, 2007