Numbers-Needed-to-Treat and Associated Costs of Care for the Treatment of Moderate-to-Severe Psoriasis

CS Hankin PhD, BioMedEcon LLC, San Jose, CA; **SR Feldman MD PhD,** Wake Forest University School of Medicine, Winston-Salem, NC;
A Szczotka PharmD, MedImpact Healthcare Systems Inc, San Diego, CA; **L Fish PharmD,** Fallon Community Health Plan, Worcester, MA; **DL Hankin MBA, BioMedEcon LLC, San Jose, CA**

BACKGROUND

Psoriasis, a skin disease characterized by scaly plaques, itching, and redness, afflicts nearly 8 million Americans.[1] Whereas the majority of psoriasis patients are diagnosed with mild disease, up to 30% satisfy criteria for moderate-to-severe psoriasis.[1]

Systemic treatment options for moderate-to-severe psoriasis are burgeoning: according to the National Psoriasis Foundation, there are at least 14 systemic treatments currently under development or in clinical trials.[2]

As the array of available systemic treatments for moderate-to-severe psoriasis proliferates, clinicians and healthcare systems will need to use informed, evidence-based methods to select the most appropriate and cost-effective treatments. This is a difficult task, given that head-to-head comparative trials are rarely available.

OBJECTIVE

To examine Numbers-Needed-to-Treat (NNT) for one patient to achieve PASI 75, and compare associated costs of care among currently approved systemic treatments (acitretin, cyclosporine, methotrexate, alefacept, efalizumab, etanercept) for moderate-to-severe psoriasis based on NNT analyses.

METHODS

An Expert Panel in dermatology, health services research and managed care conducted a systematic literature review (Medline search 1966 – March 2005) of systemic treatment efficacy for moderate-to-severe psoriasis in terms of 75% improvement from baseline in terms of the Psoriasis Area and Severity Index (PASI 75).[3] Study selection criteria were designed to minimize the introduction of bias:

- Peer-reviewed, published studies;
- Randomized, double-blind, placebo-controlled trials;
- Outcomes reported as percentage of patients achieving 75% improvement in the Psoriasis Area Severity Index (PASI 75);
- Time from treatment initiation to endpoint assessment of 8 to 14 weeks;
- Sample size in the active treatment arm of at least 10 subjects;
- Adult patients (age 18 years and above) with moderate-to-severe psoriasis or baseline PASI scores of at least 8 (signifying at least moderate severity);
- Plaque psoriasis as the predominant subtype represented;
- Unbiased subject selection (i.e., selection not based upon prior positive response to the targeted treatment);
- Dosage clearly specified;
- Treatment initiated de novo (rather than as maintenance following stabilization);
- Dosage within current product labeling.

We excluded duplicates of studies already reviewed by us, studies that did not apply an intent-to-treat analysis, where dropouts were not included in the analysis, or where the disposition of dropouts was unclear.

NNT and 95% confidence intervals (CI) were calculated using PASI 75 endpoints reported in identified studies. NNT was defined as the average number of patients who must receive a particular treatment in order for one patient to achieve PASI 75, and was calculated as the reciprocal of the percentage success achieved by the active treatment group less the percentage success achieved by placebo group.

Total annualized costs were calculated from January 2005 US Average Wholesale Drug Prices (AWP) and 2005 Medicare Reimbursement Rates (MRR) as the sum of drug, administration (e.g. IV infusion), monitoring, and risk-adjusted adverse events costs. [4,5]

Cost effectiveness, the cost required to achieve PASI 75 in one patient, was then calculated for each treatment by multiplying the NNT by total annualized treatment costs.

RESULTS

Among 2,181 articles identified from MEDLINE, 6 met selection criteria for inclusion in our calculation of NNT to achieve PASI 75 (Table 1). Table 2 shows medication regimens assumed and annualized costs of care. NNT and total annualized total costs were applied to determine total annualized costs to achieve PASI 75 in one patient (Table 2).

Table 1. NNT to Achieve PASI 75 with 95% Confidence Interval (95% CI) by Treatments and Dosage, References and Times to Endpoints

Treatment	Citation	Dose	Time to Endpoint	N Receiving Treatment	N Achieving PASI 75	N Receiving Placebo	N Achieving PASI 75	NNT (95% CI)	p-value
acitretin	Lassus [6]	50 mg/d	Week 12	20	13	20	4	2 (1 to 6)	0.011
alefacept	Lebwohl[7]	15 mg/wk	Week 14	166	55	168	22	5 (3 to 9)	<0.0001
efalizumab	Gordon[8]	1 mg/kg/wk	Week 12	369	98	187	8	4 (4 to 6)	<0.0001
	Lebwoh9	1 mg/kg/wk	Week 12	232	52	122	6	6 (4 to 9)	<0.0001
	Leonardi[10]	1mg/kg/wk	Week 12	162	63	170	4	3 (2 to 3)	<0.0001
	Combined	1mg/kg/wk	Week 12	763	213	479	18	4 (4 to 5)	<0.0001
etanercept	Gottlieb[11]	50 mg 3xs/wk	Week 12	164	81	166	6	2 (2 to 3)	<0.0001

Sensitivity Analysis. We examined uncertainty in our economic evaluation by considering the range of possible NNT values presented for each treatment within their 95% confidence intervals (CI). We considered "best guess", most conservative, and least conservative estimates by applying the NNT, lowest 95% CI bounds, and highest 95% CI bounds to costs (Figure 1).

Figure 1. Estimated Annualized Costs and 95% Confidence Intervals Required to Achieve PASI 75 for One Patient

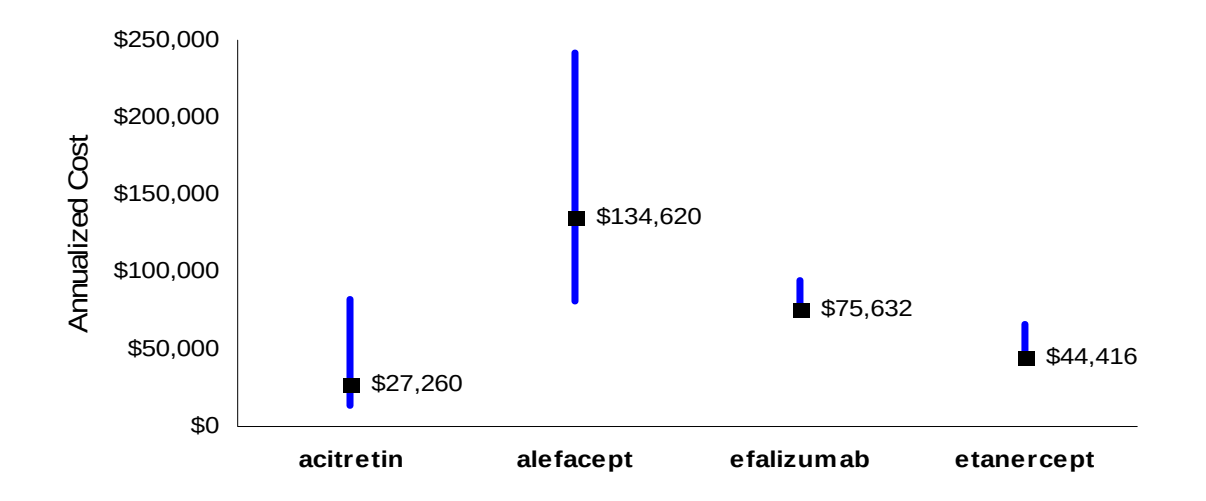
$250,000
$200,000
$150,000
$100,000
$50,000
$0
Annualized Cost
$27,260
$134,620
$75,632
$44,416
acitretin
alefacept
efalizumab
etanercept

Table 2. Medication Regimen and Annualized Costs of Care

	Annualized Costs of Care					Efficacy	Cost-Effectiveness	
Treatment	Drug Cost	Administration Cost	Monitoring Cost	Risk-Adjusted Cost of Adverse Events	TOTAL COSTS	NNT (95% CI)	Costs for One Patient to Achieve PASI 75	(95% CI)
acitretin 50 mg	$12,914	$0	$636	$80	$13,630	2 (1 to 6)	**$27,260**	($13,630 to $81,780)
alefacept 15 mg IM	$23,880	$614	$2,430	-	$26,924	5 (3 to 9)	**$134,620**	($80,772 to $242,316)
efalizumab 1 mg/kg	$18,728	$0	$180	-	$18,908	4 (4 to 5)	**$75,632**	($75,632 to $94,540)
etanercept 50 mg	$22,085	$0	$123	-	$22,208	2 (2 to 3)	**$44,416**	($44,416 to $66,624)

LIMITATIONS

- We included only peer reviewed publications (potential for publication bias).
- Efficacy in terms of PASI 75 only (an FDA-recognized endpoint).
- Regimens (administration, monitoring) per labels may not reflect actual use.
- Efficacy achieved at end of trials assumed to persist through the year.
- Results across efalizumab trials were combined (sample selection criteria, treatment interventions, study endpoints, and analytic methods were strikingly similar across trials.)
- Analysis was developed for patients with moderate-to-severe "skin" psoriasis; findings may not extend to patients with psoriatic arthritis.
- Selection criteria may have been too stringent (Other psoriasis reviews confirm poor quality in study design and presentation of results).

CONCLUSIONS

Although treatment with more expensive medications may be well-justified, decisions about *which* treatments to apply, and the order in which treatments are initiated, should take into account safety, efficacy, and costs. In the absence of head-to-head comparative trials, our methodology, which applies an evidence-based approach to cost-effectiveness analysis, represents one means of comparing safety, efficacy and costs among systemic treatments for moderate-to-severe psoriasis.

REFERENCES

1 Koo J. Dermatol Clin 1996;14:485-96. **2.** National Psoriasis Foundation Website http://www.psoriasis.org/research/ pipeline/chart.php accessed 3/16/2005. **3.** Fredriksson. Dermatologica. 1977; 157;238-244. **4.** Red Book. 2004 Edition. Montvale, NJ: Thompson Micromedex; 2004 **5.** RBRVS EZ-Fees [computer program]. Version 1.0.0. Milwaukee, WI: DMD Medical Publishers Ltd;2005. **6.** Lassus. Br J Dermatol. 1987;117:333-341. **7.** Lebwohl. Arch Dermatol. 2003;139:719-727. **8**. Gordon. JAMA 2003;290: 3073-3080. **9**. Lebwohl. N Engl J Med. 2003;20:349(21): 1987-1990.. **10**. Leonardi. J Am Acad Dermatol. 2005;52: 425-33. **11**. Gottlieb. Arch Dermatol. 2003;139:1627-1632.

Disclosures: *Dr. Hankin is a consultant for Connetics Corporation.* ***Funding Source:*** *This study was funded by Connetics Corp.*

Presented at the 10th Annual meeting of the International Society of Pharmacoeconmics and Outcomes Research, 5/05, Wash, DC.

Corresponding author: Cheryl S. Hankin, Ph.D., BioMedEcon, LLC, 2316 Walden Square, San Jose, CA 95124, 408-269-2174, cherylhankin@biomedecon.com