ESTIMATED UNITED STATES INCIDENCE AND COST OF EMERGENCY DEPARTMENT STAFF ASSAULTS PERPETRATED BY AGITATED ADULT PATIENTS WITH SCHIZOPHRENIA OR BIPOLAR DISORDER

C.S. Hankin,[1] M.M. Norris,[2] A. Bronstone[1]

BioMedEcon, Moss Beach, Calif.;[1] Mount Sinai Medical School, New York, N.Y.[2]

REVISED ABSTRACT

Objectives: To estimate the U.S. annual incidence of assaults perpetrated by agitated adult patients with schizophrenia or bipolar disorder on nurses and physicians working in general emergency departments (EDs).

Methods: A MEDLINE (1966-1/2010) and Internet search identified published reports of U.S. rates of agitation, aggression, or violence in general EDs, and/or the proportion of ED staff who reported adverse outcomes associated with patient violence (e.g., physical injury). "True" annual incidence was calculated as the average annualized number of reported physical assault cases divided by the estimated "actual" (or "true") number of assault cases believed to have occurred during the same period.

Results: Twenty-three relevant studies were identified, including staff surveys (16 studies), staff focus groups (1), retrospective analyses of staff administrative reports or police logs (5), and prospective observations (1). On average, 366 ED nurses and 226 physicians reported approximately 3.2 and 1.1 physical assaults, respectively, each year. Based on the average number of physical assaults reported each year, among a total of 31,905 ED nurses and 17,000 ED physicians practicing in the U.S., an estimated 120,796 physical assaults were perpetrated by patients. Approximately 55% of actual assaults upon staff are unreported. The "true" annual incidence of assaults perpetrated upon ED nurses and physicians by all patients appears to exceed 250,000 [(31,905 * 7.0) +(17,000 * 2.4)]. Based on reports that patients with schizophrenia and bipolar disorder are responsible for 17.1% and 11.4%, respectively, of general ED physical assaults, the "true" incidence of annual assaults upon ED nurses and physicians attributed to these patients is 75,278 [(17.1% + 11.4%) * 264,135]. On average, 1.6 and 3.5 workdays are lost by approximately one-third of ED nurses and physicians, respectively, following physical assault, at an annual cost of nearly $100 million.

Conclusion: The exact burden of staff physical assault perpetrated by agitated patients with schizophrenia or bipolar disorder remains unknown, due in part to staff tendencies toward underreporting these events. Such underreporting is unfortunate, given the substantial consequences of agitation associated with schizophrenia and bipolar disorder borne by staff, patients, caregivers, and health care systems. These consequences include more than 150,000 nursing and physician workdays lost per year, at an estimated annual cost of nearly $100 million.

BACKGROUND

- Patients with a primary psychiatric disorder diagnosis account for approximately 4.3 million (3.6%) of the estimated 119.2 million annual U.S. ED visits.[1]
- ED patients with psychiatric disorders, particularly schizophrenia and bipolar disorder, often present to U.S. EDs in an agitated state.[2]
- Clinically significant agitation (e.g., explosive and/or unpredictable anger; intimidating behavior; restlessness, pacing, or excessive movement; physical and/or verbal self-abusiveness; demeaning or hostile verbal behavior; uncooperative or demanding behavior or resistance to care; impulsive or impatient behavior; low tolerance for pain or frustration) may rapidly escalate to physical assault of ED staff.[3]
- The adverse consequences of patient physical assault on ED staff are varied (e.g., psychological response, physical injury, lost workdays, staff turnover),[4,5] and there is a dearth of U.S. research that has examined the economic impact of patient assault on ED staff.
- Although various studies have examined the incidence of assaultive behavior among U.S. ED patients and the proportion of ED staff who report having experienced patient assaultive behavior, the "true" incidence of patient assault on ED staff is difficult to determine due to:
 - Staff tendencies to underreport patient aggression and assault; and
 - Variations in study design, time frames for evaluation, and definitions of agitated and assaultive behaviors.

OBJECTIVES

- The purpose of this study is to estimate the U.S. annual incidence and costs of assaults perpetrated upon nurses and physicians who work in general emergency departments (EDs) by agitated adult patients with schizophrenia or bipolar disorder.

METHODS

- An Internet and MEDLINE (1966 through January 2010) search was conducted to identify published studies reporting:
 - Rates of agitation, aggression, or violence occurring in general EDs in the U.S.;
 - The proportion of ED staff who reported experiencing adverse outcomes associated with patient agitation (e.g., physical injury); and
 - Rates of underreporting of agitation, aggression, or violence occurring in general EDs in the U.S.
- Studies that focused exclusively on psychiatric EDs or employees of psychiatric EDs were excluded.
- For the MEDLINE search, Medical Subject Heading (MeSH) search terms were ("Emergency Services, Hospital" OR "Emergency Medicine" OR "Emergency Nursing") AND ("Psychomotor Agitation" OR "Aggression" OR "Violence" NOT "Domestic Violence").
- Physical assault was defined as being hit, slapped, kicked, pushed, choked, grabbed, sexually assaulted, or otherwise subjected to physical contact intended to injure or harm.
- To estimate the annual incidence of patient assaults on ED nurses and physicians, only studies examining the incidence of assaults over a one-year period were included.
- For studies that reported the number of staff who experienced physical assaults by frequency category (e.g., 1-5, 6-10), the median number of assaults within each category and the number of staff were used to calculate the total number of assaults experienced.
- The "true" annual incidence was calculated as the average annual number of assaults reported divided by the estimated "actual" (or "true") number of assault cases believed to have occurred during the same period.
 - For example, if 100 assaults were reported, and these reports were believed to represent only 10% of the "true number" of assaults, then the estimated "true" annual incidence was calculated as 100/10%, or 1,000.

RESULTS

Description of included studies

- A total of 23 studies were identified as relevant to the study objectives. Characteristics of these studies are summarized in Table 1.

Table 1. Characteristics of Included Studies

Study	Design	Population	Setting
Barlow, 1997[6]	Cross-sectional mailed survey	Convenience sample of 475 surgical residents from 57 of 206 residency programs	57 university-affiliated general EDs
Behnam, 2010[7]	Cross-sectional online survey	Convenience sample of 263 ED residents	65 randomly selected (out of 134) accredited U.S. emergency medicine residency programs
Black, 1994[8]	Cross-sectional mailed survey	42 general psychiatry residents recruited from a convenience sample of 42	3 university-affiliated general EDs (2 private, 1 public)
Erickson, 2000[9]	Cross-sectional mailed survey	2 general EDs	Convenience sample of 55 nurses
Foust, 1993[10]	9-month, prospective, observational study	48,938 consecutive ED patients	1 university-affiliated general ED Level I trauma center with annual census of ~64,000
Gacki-Smith, 2009[11]	Cross-sectional online survey	3,465 ED nurses from a convenience sample of 31,905 Emergency Nurses Association members	EDs throughout the 50 states and D.C.
Ganzini, 1995[12]	5-year retrospective review of staff incident reports and patient medical records	57,791 consecutive ED patient visits	1 Department of Veterans Affairs Medical Center general ED
Gates, 2006[13]	Cross-sectional mailed survey	242 ED employees from a convenience sample of 600	5 EDs (4 general, 1 Level I trauma center with separate medical, psychiatric, and air care EDs)
Gerberich, 2004[5]	Cross-sectional mailed survey	4,918 licensed nurses (115 working in EDs) from a random sample of 6,800	Various general EDs in MN
Kasangra, 2008[14]	Cross-sectional mailed survey	3,518 ED staff from a convenience sample of 5,695	65 of 69 general EDs in MI
Kowalenko, 2005[15]	Cross-sectional mailed survey	177 ED physicians from a random sample of 250	Various general EDs in MI
Lavoie, 1988[16]	Cross-sectional mailed survey	127 ED medical directors from a convenience sample of 170	University-affiliated general EDs with ≥40,000 annual patient visits
Levin, 1988[4]	Cross-sectional focus group	Convenience sample of 22 ED nurses	15 general EDs
Li, 2010[17]	Cross-sectional mailed survey	196 emergency medicine residents from a convenience sample of 380	10 emergency medicine programs
Mahoney, 1991[18]	Cross-sectional mailed survey	1,209 ED nurses from a convenience sample of ~2,000	124 of 222 general EDs in PA
May, 2002[19]	Cross-sectional hand-delivered survey	86 RNs (28 ED RNs) from a convenience sample of 125	1 general ED in FL
Mayer, 1999[20]	Cross-sectional mailed survey	226 ED personnel from a convenience sample of 600	18 of 19 general EDs in FL
Ordog, 1995[21]	14-year retrospective review of ED security records	980,000 consecutive ED patients	1 urban general ED
Pane, 1991[22]	1-year retrospective review of university police logs and ED staff incident reports	693 episodes of disturbance or violence	1 university-affiliated general ED Level I trauma center with annual census of ~40,000
Peek-Asa, 2002[23]	Cross-sectional mailed survey	95 ED nurse managers from a convenience sample of 180	180 general EDs in CA
Peek-Asa, 2007[24]	10-year retrospective review of various sources of information regarding workplace violence	Convenience sample of 100 ED RNs	50 general EDs in NJ
Presley, 2002[25]	Cross-sectional mailed survey	Convenience sample of 101 RNs working in general EDs	General EDs in Austin, TX
Rankins, 1999[26]	29-month retrospective study of security department records	155,976 consecutive ED patients	1 urban general ED

Figure 1. Percentage of ED Staff Experiencing Patient Physical Assault

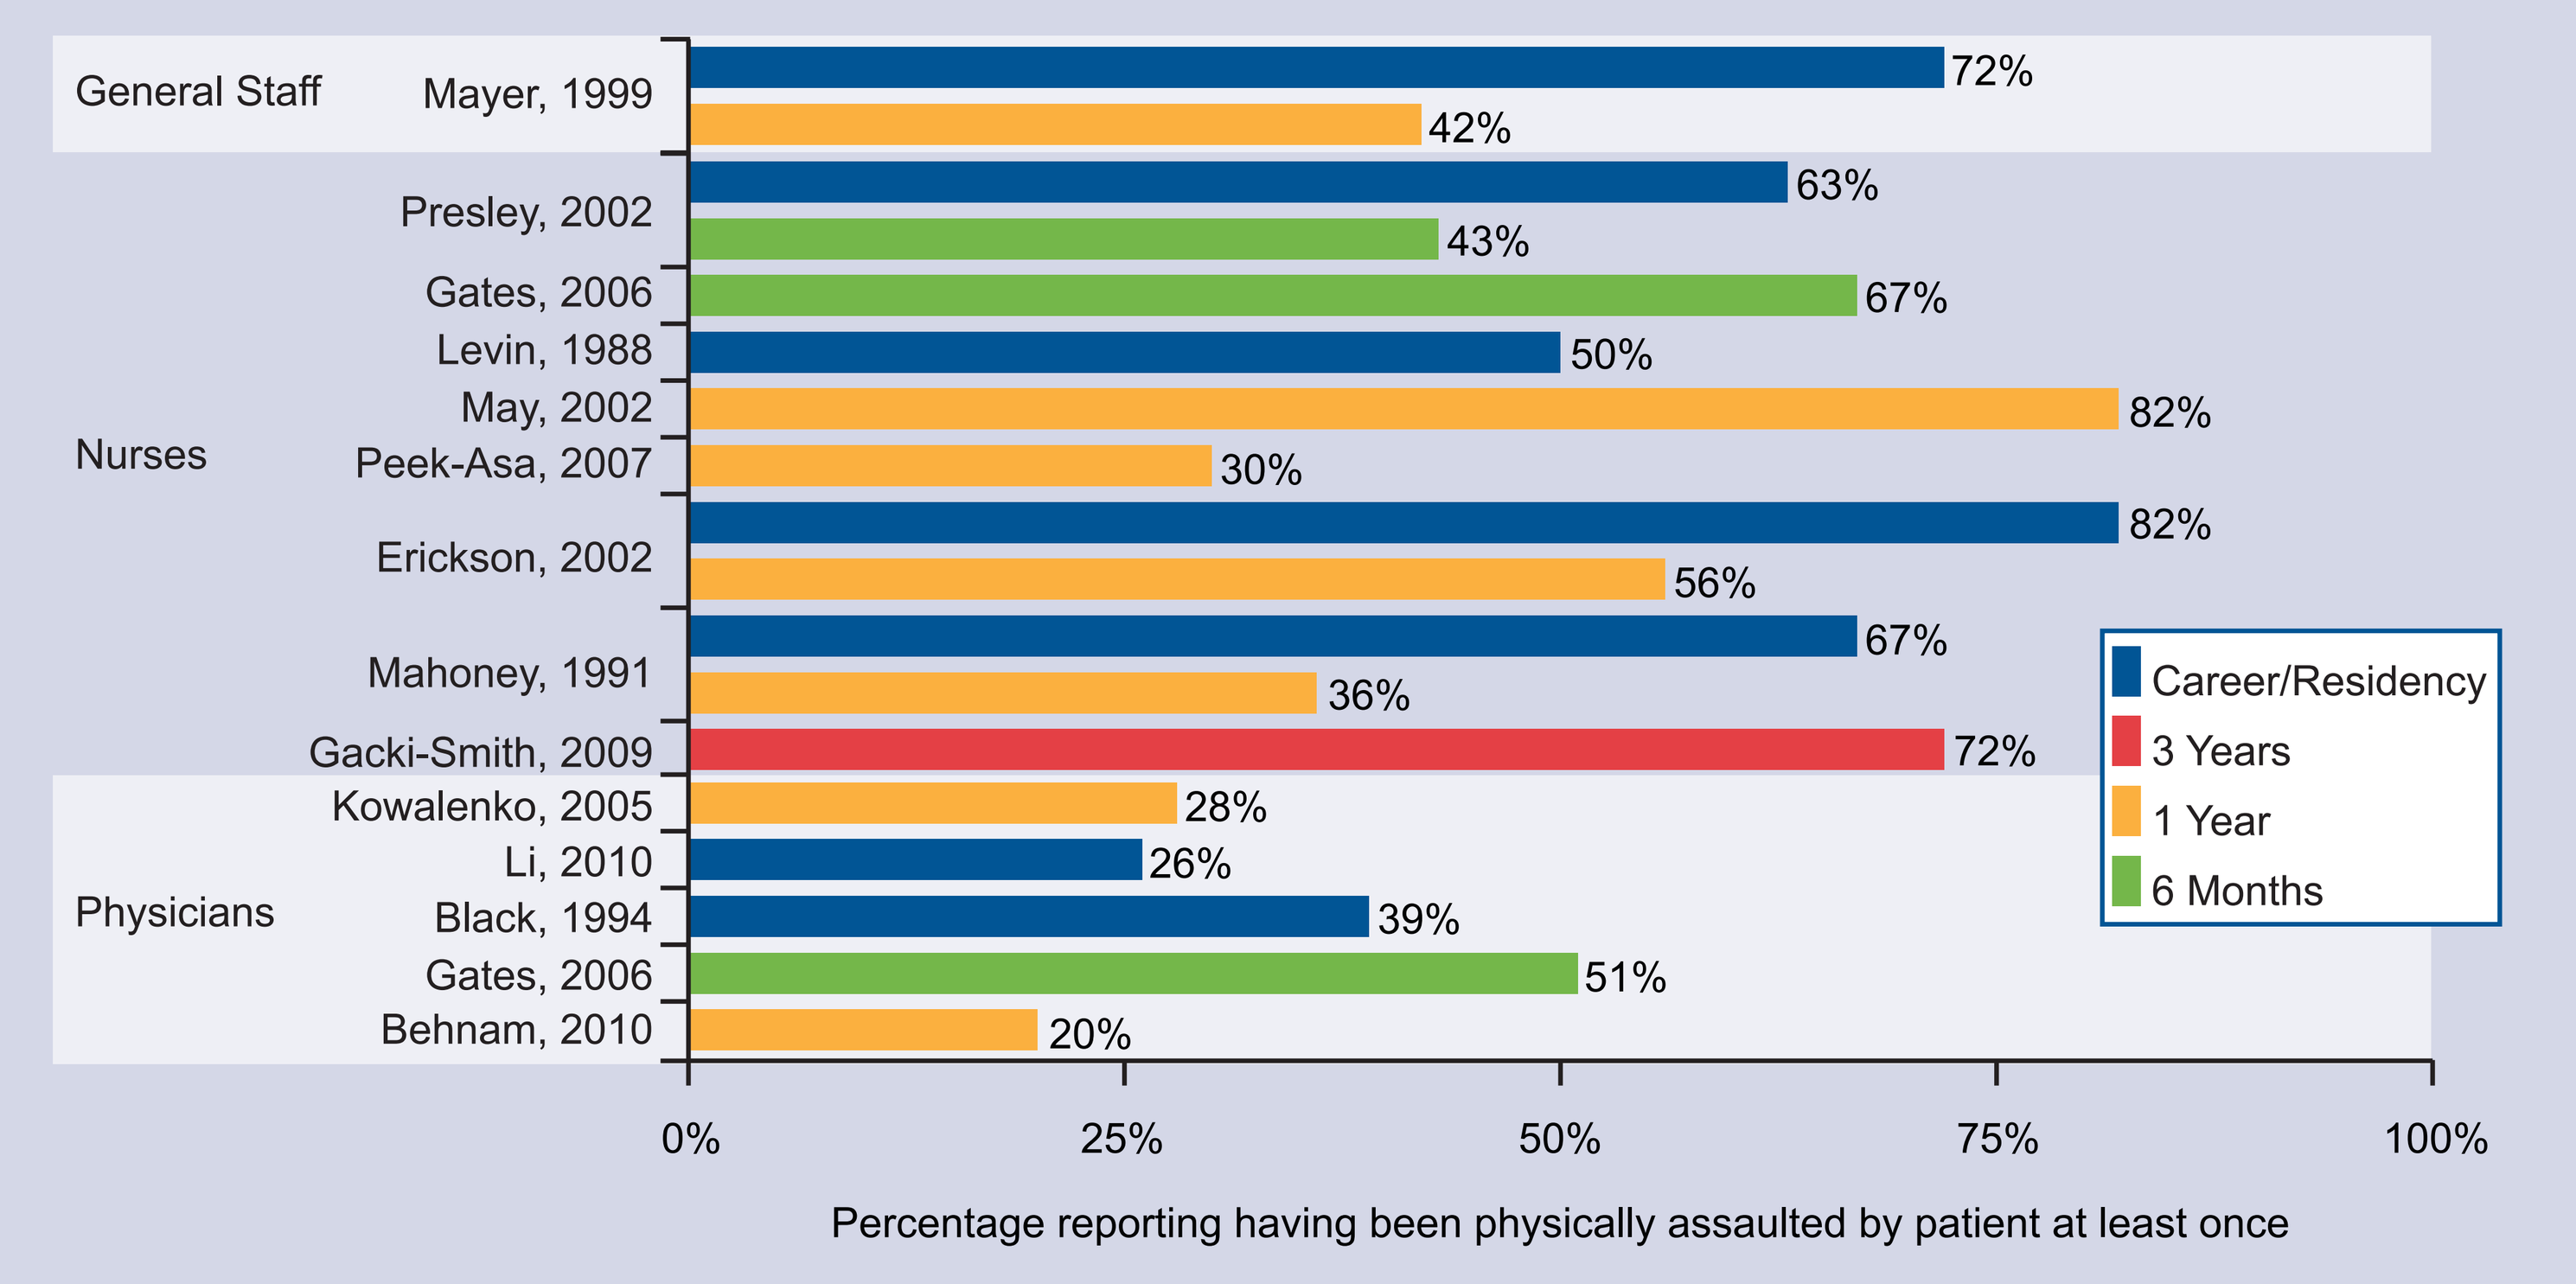
General Staff
Mayer, 1999
72%
42%
Nurses
Presley, 2002
63%
43%
Gates, 2006
67%
Levin, 1988
50%
May, 2002
82%
Peek-Asa, 2007
30%
Erickson, 2002
82%
56%
Mahoney, 1991
67%
36%
Gacki-Smith, 2009
72%
Physicians
Kowalenko, 2005
28%
Li, 2010
26%
Black, 1994
39%
Gates, 2006
51%
Behnam, 2010
20%
0%
25%
50%
75%
100%
Percentage reporting having been physically assaulted by patient at least once
Career/Residency
3 Years
1 Year
6 Months

Commonly cited reasons for ED staff assaults by patients[9,10,19]

- **Patient Factors**
 - Psychiatric disorders
 - Alcohol/drug use
 - Organic brain syndrome/dementia
- **Staff Factors**
 - Insufficient staffing
 - Lack of violence prevention training
 - Lack of information regarding patients' violence history
- **Environmental Factors**
 - Long wait times
 - Use of restraint/seclusion
 - Lack of security presence
 - Patient areas and triage open to public
 - Poor security response/not helpful
 - Ability of public to bring weapons into ED
 - Lack of metal detectors
 - Lack of procedures/policies for handling known violent offenders

Table 2. Underreporting of Physical Assault

Study	% Underreporting* of Physical Assault
Behnam, 2010[8]	15%
Erickson, 2002[9]	29%
Foust, 1993[10]	79%
Gates, 2006[13]	65%
Gerberich, 2004[5]	60%
May, 2002[19]	50%
Peek-Asa, 2007[23]	72%
Presley, 2002[25]	70%
Average	**55%**

*Events that were not formally reported in writing were considered unreported.

On average, 55% of physical assaults by patients are not reported.

Table 3. Rates of Annual Physical Assaults on ED Nurses and Physicians by Patients

Study	Number of General ED Nurses and Physicians	Number of Reported Annual Physical Assaults	("True") Estimated Annual Incidence of Physical Assaults [Reported Number of Physical Assaults/45%]	Average Annual Incidence of Reported Physical Assaults per ED Nurse or Physician	Average Adjusted ("True") Annual Incidence of Reported Physical Assaults per ED Nurse or Physician
Nurses					
Gates, 2006[13]	95	251	558		
Mayer, 1999[20]	171	781	1,736		
Peek-Asa, 2007[23]	100	121	269		
TOTAL	**366**	**1,153**	**2,563**	**1,153/366 = 3.2**	**2,563/366 = 7.0**
Physicians					
Gates, 2006[13]	49	130	289		
Kowalenko, 2005[15]	177	115	256		
TOTAL	**226**	**245**	**545**	**245/226 = 1.1**	**545/226 = 2.4**

*Assumes that 55% of physical assaults are not reported (see Table 2)

- Assuming that there are 31,905 ED nurses[27] and 17,000 board-certified ED physicians[28] currently practicing in the U.S., the total number of reported annual physical assaults perpetrated by patients on staff in the general ED is 120,796. [(31,905 * 3.2] + (17,000 * 1.1)] = [102,096 assaults on nurses + 18,700 assaults on physicians]
- Given that an estimated 55% of physical assaults are unreported, the "true" total number of annual assaults is estimated to be 264,135. [(31,905 * 7.0) + (17,000 * 2.4)] = [223,335 assaults on nurses + 40,800 assaults on physicians annually]
- Patients with schizophrenia and bipolar disorder are reportedly responsible for 17.1% and 11.4%, respectively, of physically assaultive behaviors in the ED[12] and thus may account for 75,278 [(17.1% + 11.4%) * 264,135] annual physical assaults perpetrated upon nurses and physicians in the general ED (63,650 assaults on nurses and 11,628 assaults on physicians).

Feelings of safety among ED staff

- **"How often do you feel safe from violence when working in the ED?"[13]**
 - 6% "Never"
 - 12% "Seldom"
 - 21% "Occasionally"
 - 54% "Often"
 - 7% "Always"
- **"Overall, how safe do you feel from patient physical assaults?"[9]**
 - 7% "None of the time"
 - 27% "Some of the time"
 - 62% "Most of the time"
 - 4% "All of the time"
- **"How often are you fearful of workplace violence?"[15]**
 - 1% "Constantly fearful"
 - 9% "Frequently fearful"
 - 82% "Occasionally fearful"
 - 8% "Never fearful"
- **"How often have you felt physically safe while working in the ED?"[14]**
 - 73% "Most of the time or always"
 - 19% "Sometimes"
 - 8% "Rarely or never"

Annual cost of missed work due to patient assault on ED nurses and physicians

- In a 10-year retrospective study of 100 EDs in New Jersey, approximately one-third (33%) of assaulted employees missed at least one full day of work as a result of the incident.[23]
- In a one-year retrospective study of 8,552 incidents involving assaults reported by 166 Department of Veterans Affairs Medical Center facilities, 1,085 injured nurses and 46 injured physicians incurred a total of 1,734 and 163 lost workdays, respectively.[29]
 - Injured nurses lost an average of 1.6 (1,734/1,085) workdays
 - Injured physicians lost an average of 3.5 (163/36) workdays
- As previously noted, assuming a total of 264,135 annual U.S. assaults in the general ED perpetrated by patients upon staff, an estimated
 - 223,335 will be experienced by ED nurses, of which an estimated 73,700 (33%) assaults will result in 117,921 lost workdays (1.6 nursing days per assault)
 - 40,800 will be experienced by ED physicians, of which an estimated 13,464 (33%) assaults will result in 47,124 lost workdays (3.5 physician days per assault)
- Recent hourly pay rates with benefits reported for nurses and doctors are $43 and $147,* respectively. The total estimated annual cost of staff workdays lost due to patient physical assault in the general ED is $96 million.
 - ED Nurses: $43 hourly rate * 117,921 days * 8 hours/day = $40.6 million
 - ED Physicians: $147 hourly rate * 47,124 days * 8 hours/day = $55.4 million

*Average hourly salary for ED nurse (RN) and physician in the U.S. based on a national median annual salary (base, benefits, and bonus) of $88,507 and $304,942, respectively (assumes 40-hour workweek and 52-week work year).
Data obtained from Salary.com. Salary Wizard. Accessed May 8, 2010. Available at: http://swz.salary.com/salarywizard/layoutscripts/swzl_selectjob.asp?op=salswz_psr&hdSearchByOption=0&hdLocationOption=0&hdKeyword=nurses+emergency&hdJobCategory=&hdZipCode=&hdStateMetro=&hdNarrowDesc=&rdbSearchByOption=0&txtKeyword=nurses+emergency&rdbLocationOption=0&txtZipCode=.

CONCLUSION

- General EDs in the U.S. are overwhelmed by a growing proportion of visits due to mental illness (a 28% increase from 1992-2001[31]) and a decrease in the number of available psychiatric and substance abuse treatment resources.[32-33]
- ED patients with a primary psychiatric diagnosis often present as agitated,[2] and there are an inadequate number of ED staff with psychiatric expertise.[34]
- Agitation encompasses a wide range of behaviors that typically occur along a continuum, escalate in a predictive manner and, in the absence of timely and effective intervention, may culminate in the physical assault of staff.[35]
- Although we identified 23 studies that reported information regarding physical assault of ED staff by patients, only four studies provided sufficient information for estimating the annual incidence of physical assaults experienced by U.S. nurses and physicians in the ED.
- Eight studies noted substantial underreporting of these incidents. On average, over half (55%) of physical assaults upon staff were not reported.
- Nurses and physicians who provide care in general EDs will likely treat greater numbers of patients with mental disorders in the future, as community mental health resources continue to dwindle. ED staff commonly lack specialized training and effective means to treat patients with schizophrenia or bipolar disorder who present with agitation, thereby increasing the likelihood for staff physical assault by agitated patients. The exact burden of staff physical assault perpetrated by agitated patients with schizophrenia or bipolar disorder remains unknown, due in some part to staff tendencies toward underreporting these events.
- Such underreporting is unfortunate, given that the consequences of agitation associated with schizophrenia and bipolar disorder borne by staff, patients, caregivers, and health care systems are substantial. These consequences include more than 150,000 nursing and physician workdays lost per year, at an estimated annual cost of $100 million. As fewer community mental health resources become available, patients with schizophrenia and bipolar disorder will increasingly seek care in the general ED. Because these patients often present with agitation that may quickly escalate to physical assault, and because agitation is associated with considerable staff, patient, caregiver, and health care systems burden, effective and timely treatment is imperative.

REFERENCES

1. Pitts SR, Niska RW, Xu J, Burt CW. National Hospital Ambulatory Medical Care Survey: 2006 emergency department summary. *Natl Health Stat Report* 2008;(7):1-38.
2. Zun LS, Downey LV. Level of agitation of psychiatric patients presenting to an emergency department. *Prim Care Companion J Clin Psychiatry* 2008;10(2):108-113.
3. Allen MH, Currier GW, Carpenter D, Ross RW, Docherty JP. The expert consensus guideline series. Treatment of behavioral emergencies 2005. *J Psychiatr Pract* 2005;11 Suppl 1:5-108.
4. Levin PF, Hewitt JB, Misner ST. Insights of nurses about assault in hospital-based emergency departments. *Image J Nurs Sch* 1998;30(3):249-254.
5. Gerberich SG, Church TR, McGovern PM, et al. An epidemiological study of the magnitude and consequences of work related violence: the Minnesota Nurses' Study. *Occup Environ Med* 2004;61(6):495-503.
6. Barlow CB, Rizzo AG. Violence against surgical residents. *West J Med* 1997;167:74-78.
7. Behnam M, Tillotson RD, Davis SM, Hobbs GR. Violence in the emergency department: A national survey of emergency medicine residents and attending physicians. *J Emerg Med* 2010.
8. Black KJ, Compton WM, Wetzel M, et al. Assaults by patients on psychiatric residents at three training sites. *Hosp Community Psychiatry* 1994;45(7):706-710.
9. Erickson L, Williams-Evans SA. Attitudes of emergency nurses regarding patient assaults. *J Emerg Nurs* 2000;26(3):210-215.
10. Foust D, Rhee KJ. The incidence of battery in an urban emergency department. *Ann Emerg Med* 1993;22(3):583-585.
11. Gacki-Smith J, Juarez AM, Boyett L, et al. Violence against nurses working in US emergency departments. *J Nurs Adm* 2009;39(7-8):340-349.
12. Ganzini L, Edwards P, Surkan PJ, Drummond DJ. Characteristics of violent elderly in the emergency department. *Int J Geriatr Psychiatry* 1995;10:945-950.
13. Gates DM, Ross CS, McQueen L. Violence against emergency department workers. *J Emerg Med* 2006;31(3):331-337.
14. Kansagra SM, Rao SR, Sullivan AF, et al. A survey of workplace violence across 65 U.S. emergency departments. *Acad Emerg Med* 2008;15(12):1268-1274.
15. Kowalenko T, Walters BL, Khare RK, Compton S. Workplace violence: a survey of emergency physicians in the state of Michigan. *Ann Emerg Med* 2005;46(2):142-147.
16. Lavoie FW, Carter GL, Danzl DF, Berg RL. Emergency department violence in United States teaching hospitals. *Ann Emerg Med* 1988;17(11):1227-1233.
17. Li SF, Grant K, Bhoj T, et al. Resident experience of abuse and harassment in emergency medicine: ten years later. *J Emerg Med* 2010;38(2):248-252.
18. Mahoney BS. The extent, nature, and response to victimization of emergency nurses in Pennsylvania. *J Emerg Nurs* 1991;17(5):282-291; discussion 292-284.
19. May DD, Grubbs LM. The extent, nature, and precipitating factors of nurse assault among three groups of registered nurses in a regional medical center. *J Emerg Nurs* 2002;28(1):11-17.
20. Mayer BW, Smith FB, King CA. Factors associated with victimization of personnel in emergency departments. *J Emerg Nurs* 1999;25(5):361-366.
21. Ordog GJ, Wasserberger J, Ordog C, Ackroyd G, Atluri S. Violence and general security in the emergency department. *Acad Emerg Med* 1995;2(2):151-154.
22. Pane GA. Preventing and controlling violence in emergency departments. *West J Med* 1991;155(3):285-286.
23. Peek-Asa C, Cubbin L, Hubbell K. Violent events and security programs in California Emergency Departments before and after the 1993 Hospital Security Act. *J Emerg Nurs* 2002;28(5):420-426.
24. Peek-Asa C, Allareddy V, Valiante D, et al. Workplace violence and prevention in New Jersey hospital emergency departments: Summary report of 50 New Jersey hospitals participating in the evaluation of California initiatives to reduce violence against healthcare workers study. Trenton, NJ: New Jersey Department of Health and Human Services; 2007.
25. Presley D, Robinson G. Violence in the emergency department: nurses contend with prevention in the healthcare arena. *Nurs Clin North Am* 2002;37(1):161-169, viii-ix.
26. Rankins RC, Hendey GW. Effect of a security system on violent incidents and hidden weapons in the emergency department. *Ann Emerg Med* 1999;33(6):676-679.
27. Available at: http://www.state.nj.us/health/eoh/survweb/documents/njhospsec_rpt.pdf.
28. Available at: http://health.howstuffworks.com/emergency-room5.htm.
29. Lehmann LS, McCormick RA, Kizer KW. A survey of assaultive behavior in Veterans Health Administration facilities. *Psychiatr Serv* 1999;50(3):384-389.
30. www.salary.com. Accessed May 6, 2010.
31. Larkin GL, Claassen CA, Emond JA, Pelletier AJ, Camargo CA. Trends in U.S. emergency department visits for mental health conditions, 1992 to 2001. *Psychiatr Serv* 2005;56(6):671-677.
32. National Alliance on Mental Illness. Emergency Departments See Dramatic Increase in People with Mental Illness Seeking Care. Available at: http://www.nami.org/Template.cfm?Section=Press_Release_Archive&template=/contentmanagement/contentdisplay.cfm&ContentID=85724&title=Emergency%20Departments%20See%20Dramatic%20Increase%20in%20People%20with%20Mental%20Illness%20Seeking%20Care. Accessed October 30, 2009.
33. American College of Emergency Physicians. ACEP Psychiatric and Substance Abuse Survey 2008. Available at: http://www.acep.org/uploadedFiles/ACEP/Advocacy/federal_issues/PsychiatricBoarding-Summary.pdf.
34. O'Malley AS, Gerland AM, Pham HH, Berenson RA. Rising pressure: hospital emergency departments as barometers of the health care system. *Issue Brief Cent Stud Health Syst Change* 2005(101):1-4.
35. Morrison EF. A hierarchy of aggressive and violent behaviors among psychiatric inpatients. *Hosp Community Psychiatry* 1992;43(5):505-506.

Funding for this research was provided by Alexza Pharmaceuticals, Inc.
For more information, please contact Cheryl Hankin, chankin@biomedecon.com, 650 563 9475.

Presented at the 15th Annual International Meeting of the International Society for Pharmacoeconomics and Outcomes Research (ISPOR); May 15-19, 2010; Atlanta, Ga