459

Adherence to Monthly Injectable Leuprolide Acetate among Florida Medicaid-enrolled Children with Central Precocious Puberty *(P459E)*

Cheryl Hankin[1], Amy Bronstone[1], Zhaohui Wang[1]

[1] BioMedEcon, LLC, Moss Beach, CA

BACKGROUND

- **Central precocious puberty (CPP)** is a rare disorder caused by premature activation of the hypothalamus-pituitary-gonadal (HPG) axis, causing development of secondary sexual characteristics, accelerated growth, and accelerated bone maturation.[1]
- **Untreated CPP is associated with:**
 - Psychosocial and behavioral problems[2-5] (e.g., early sexual behavior, early pregnancy, substance abuse, depression, conduct problems).
 - Early preemptive growth spurt[6] (early closure of epiphyseal growth plates, compromised adult stature).
- **Treatment**
 - Gonadotropin releasing hormone (GnRH) agonists are the mainstay treatment.
 - GnRH initiation causes an initial paradoxical stimulatory effect (“flare up”) that remits following continuous administration.
 - Consequences of interrupted therapy, which can lead to multiple flare ups, may include returned pubertal signs (e.g., menses, breast development, testicular growth, and compromise of adult stature).
 - In the U.S., the most commonly used GnRH agonist is an intramuscular injection of depot leuprolide acetate given once every 28 days ("monthly depot leuprolide").

Efficacy is dependent upon adherence to a 28-day regimen of monthly depot leuprolide regimen

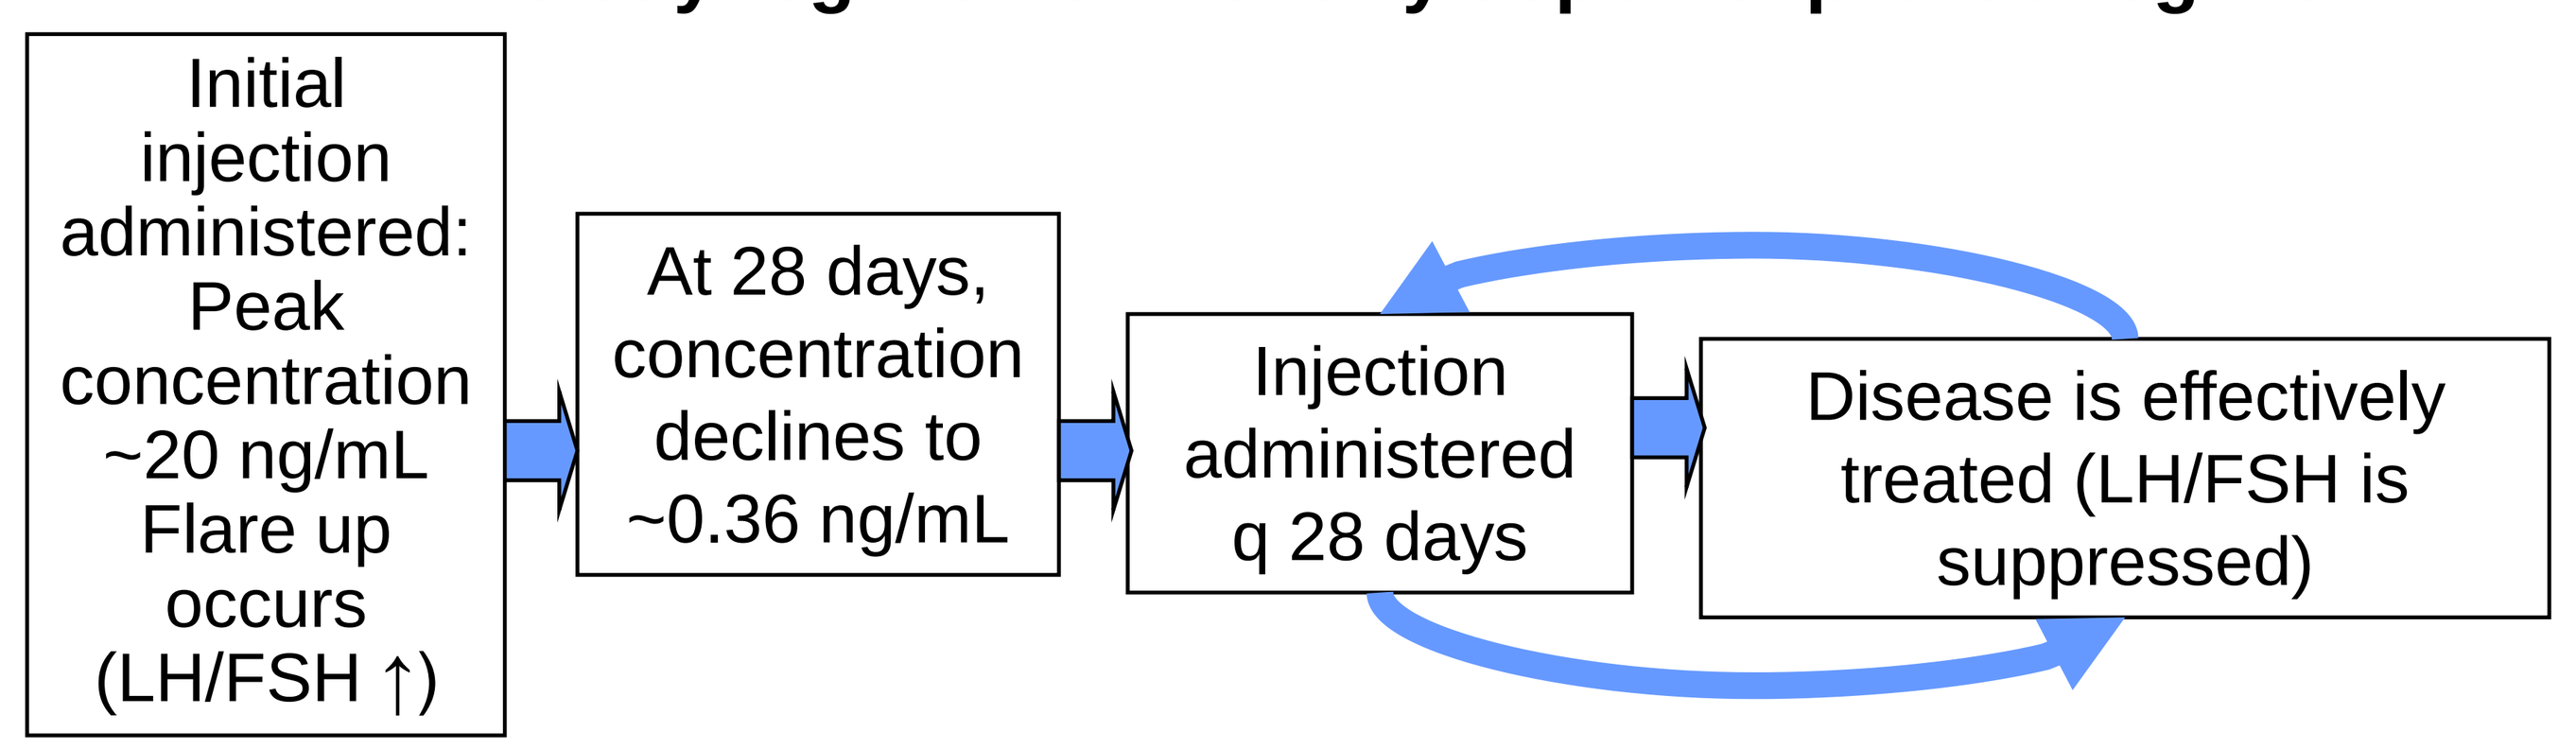
Initial injection administered: Peak concentration ~20 ng/mL Flare up occurs (LH/FSH ↑)
At 28 days, concentration declines to ~0.36 ng/mL
Injection administered q 28 days
Disease is effectively treated (LH/FSH is suppressed)

<u>Exacerbation</u> of disease occurs when patients do not adhere to a 28-day regimen of monthly depot leuprolide

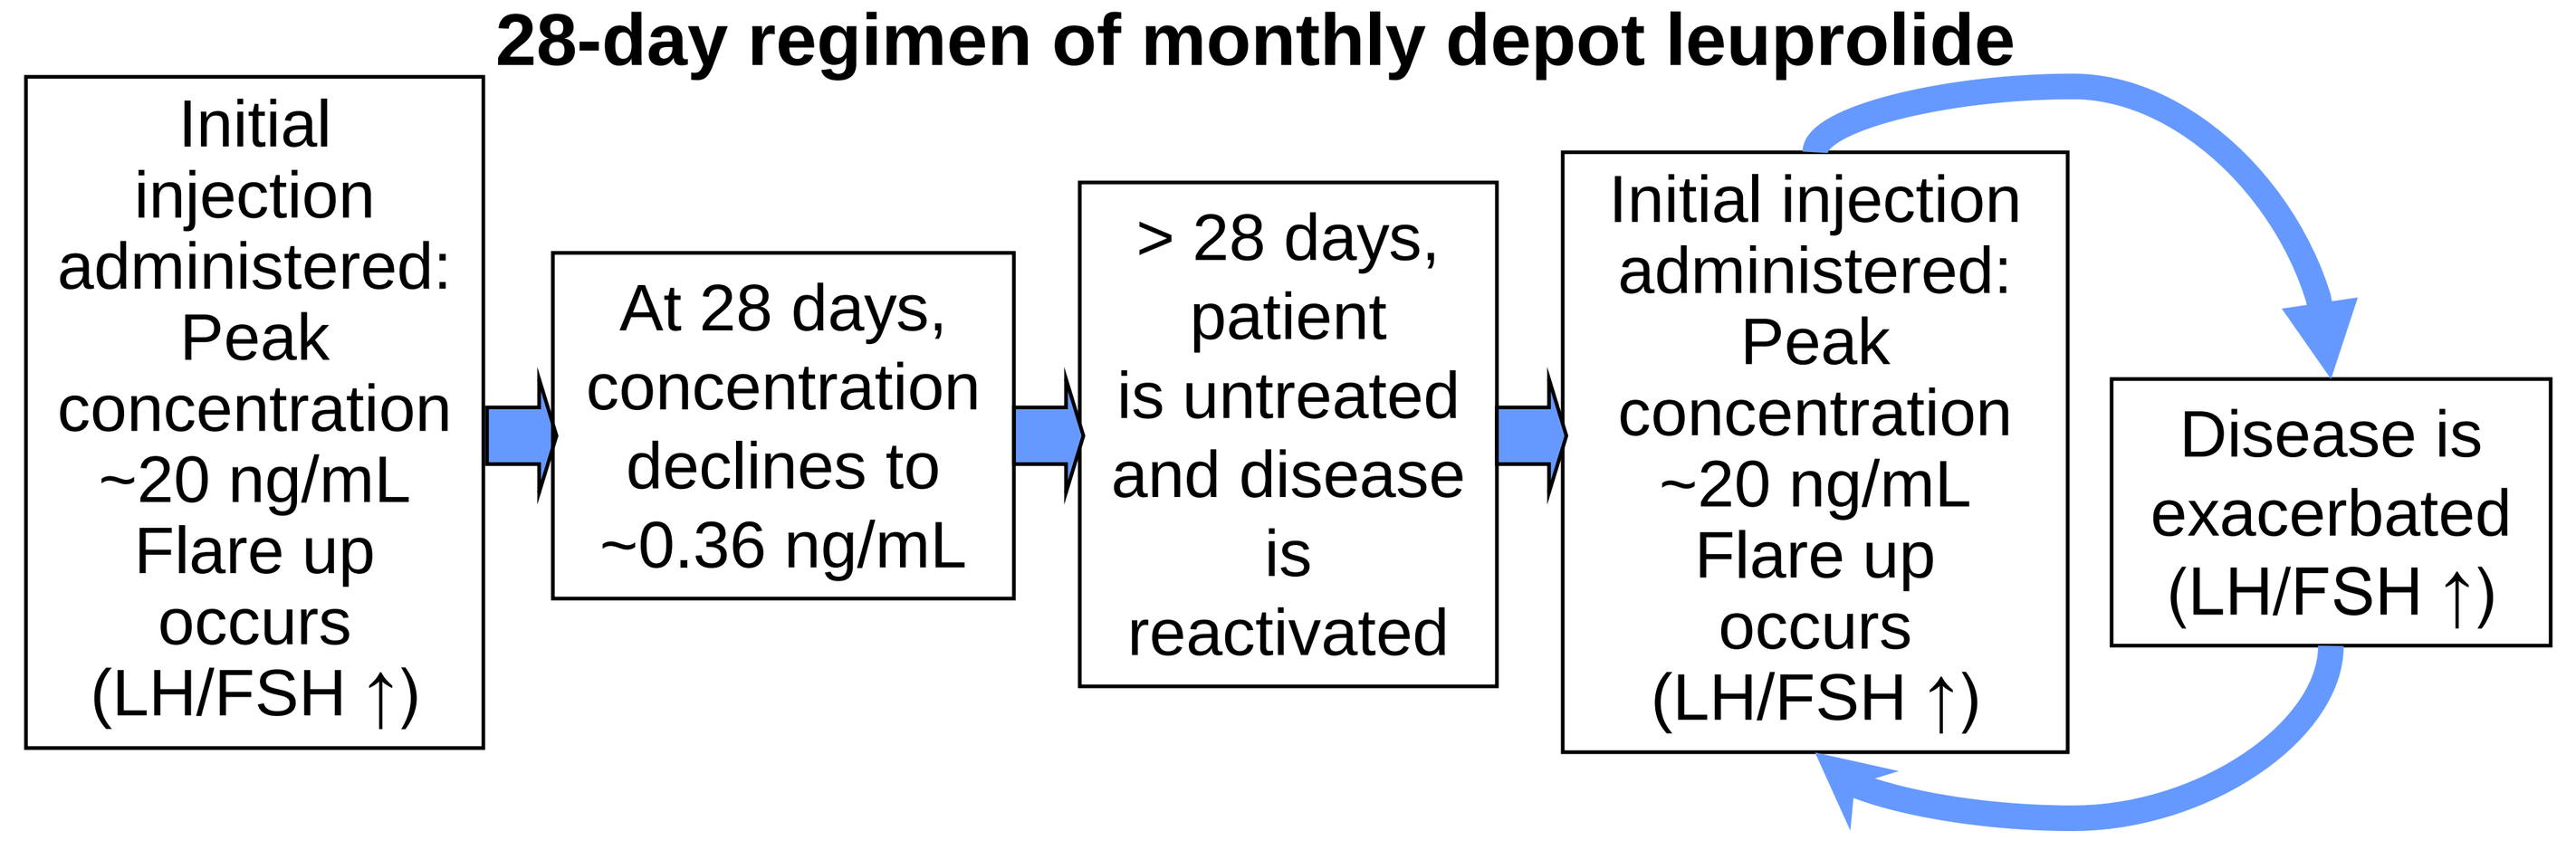
Initial injection administered: Peak concentration ~20 ng/mL Flare up occurs (LH/FSH ↑)
At 28 days, concentration declines to ~0.36 ng/mL
> 28 days, patient is untreated and disease is reactivated
Initial injection administered: Peak concentration ~20 ng/mL Flare up occurs (LH/FSH ↑)
Disease is exacerbated (LH/FSH ↑)

OBJECTIVE

- Because complete suppression of hormones is dependent on continuous delivery of GnRH agonists, we examined the extent to which patients adhered to the prescribed monthly depot leuprolide regimen.

METHODS

- We conducted a 7-year (1997-2004) retrospective analysis of Florida Medicaid-enrolled children age 2-11 years with CPP (ICD-9 259.1, precocious sexual development and puberty, including premature thelarche and adrenarche) to examine the frequency with which children received monthly depot leuprolide. Patients who received at least one administration of monthly depot leuprolide were identified. All data were patient de-identified and HIPAA compliant.

RESULTS

Among 3,530 children with CPP, 266 (7.5%) received monthly depot leuprolide administrations during the index period.

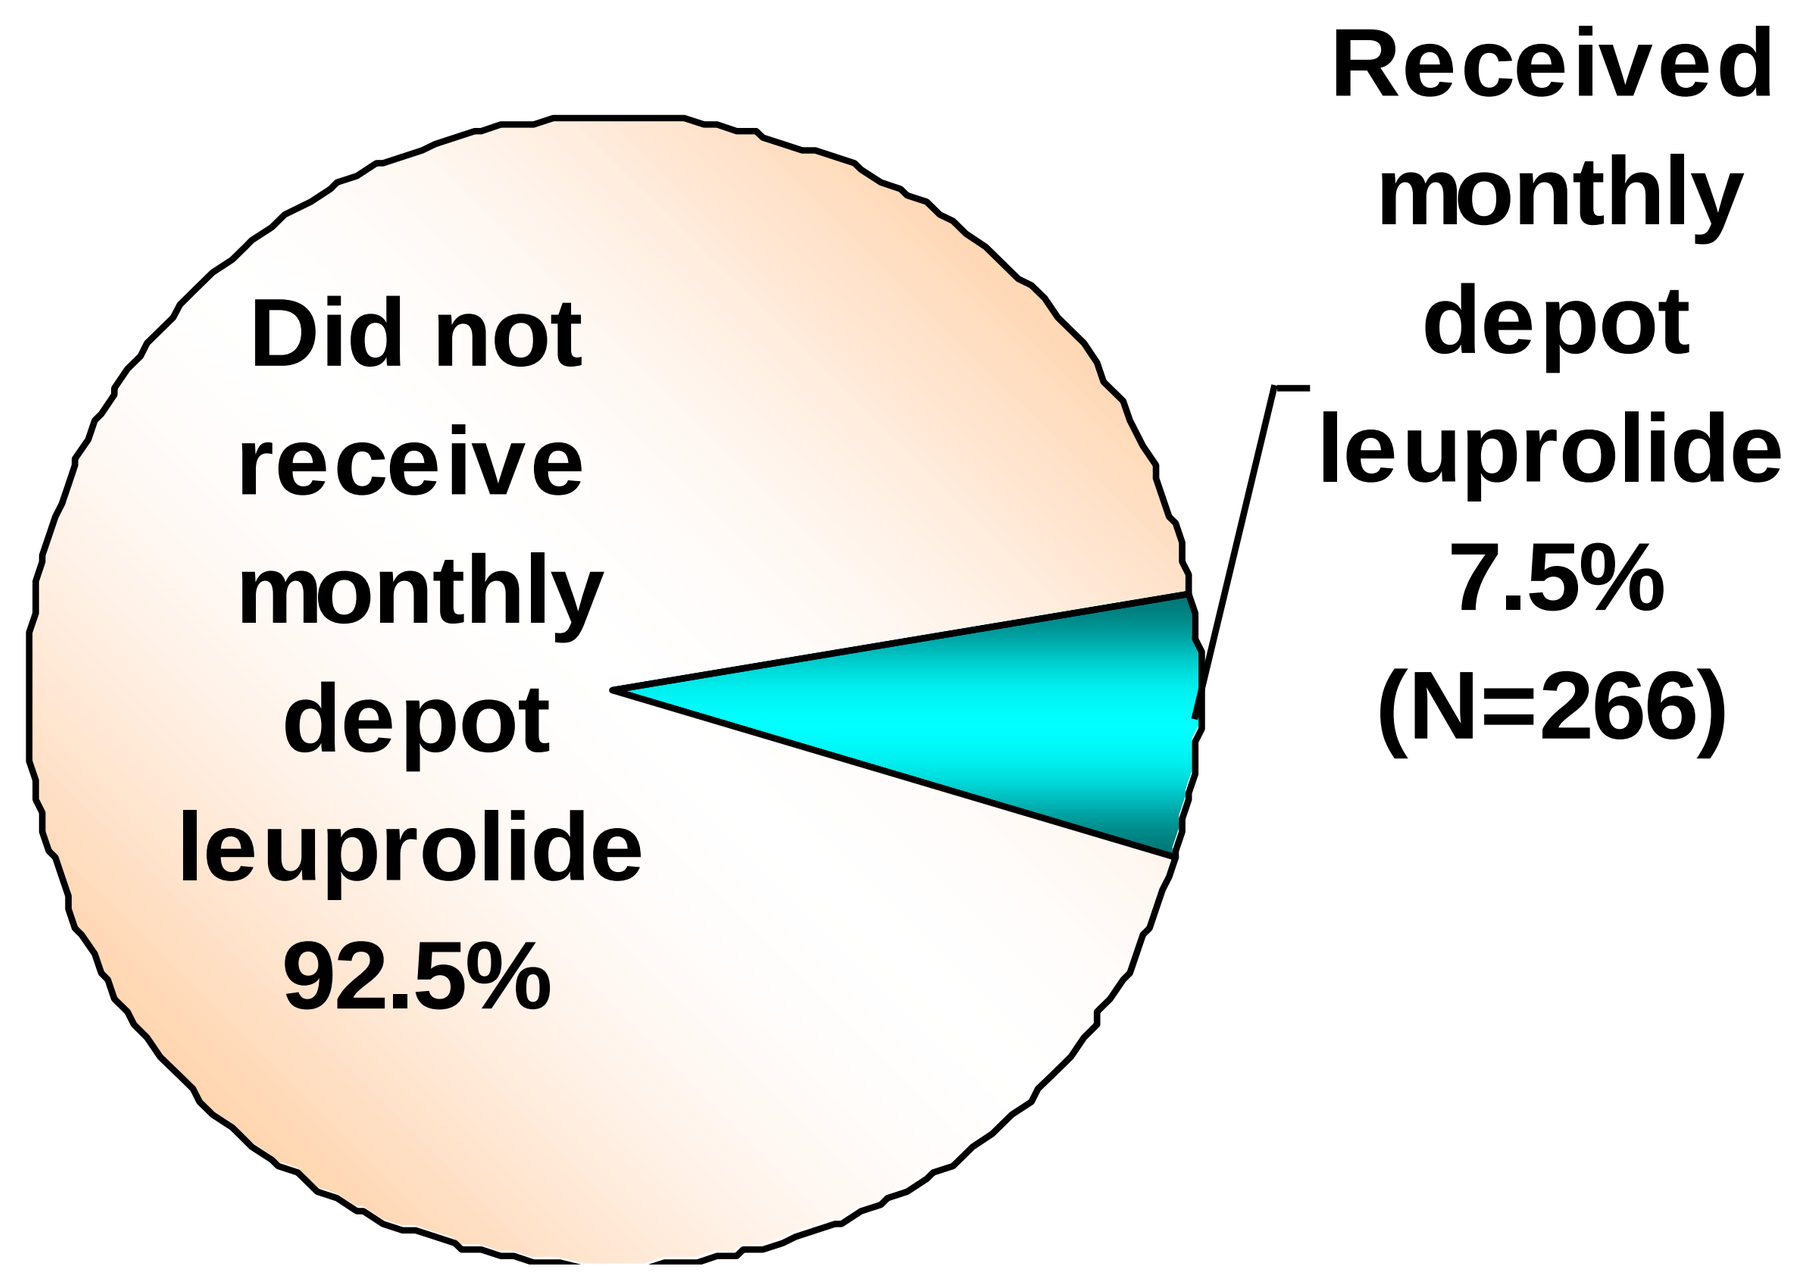
Did not receive monthly depot leuprolide 92.5%
Received monthly depot leuprolide 7.5% (N=266)

Sex distribution among those receiving monthly depot leuprolide administrations during the index period (N=266)

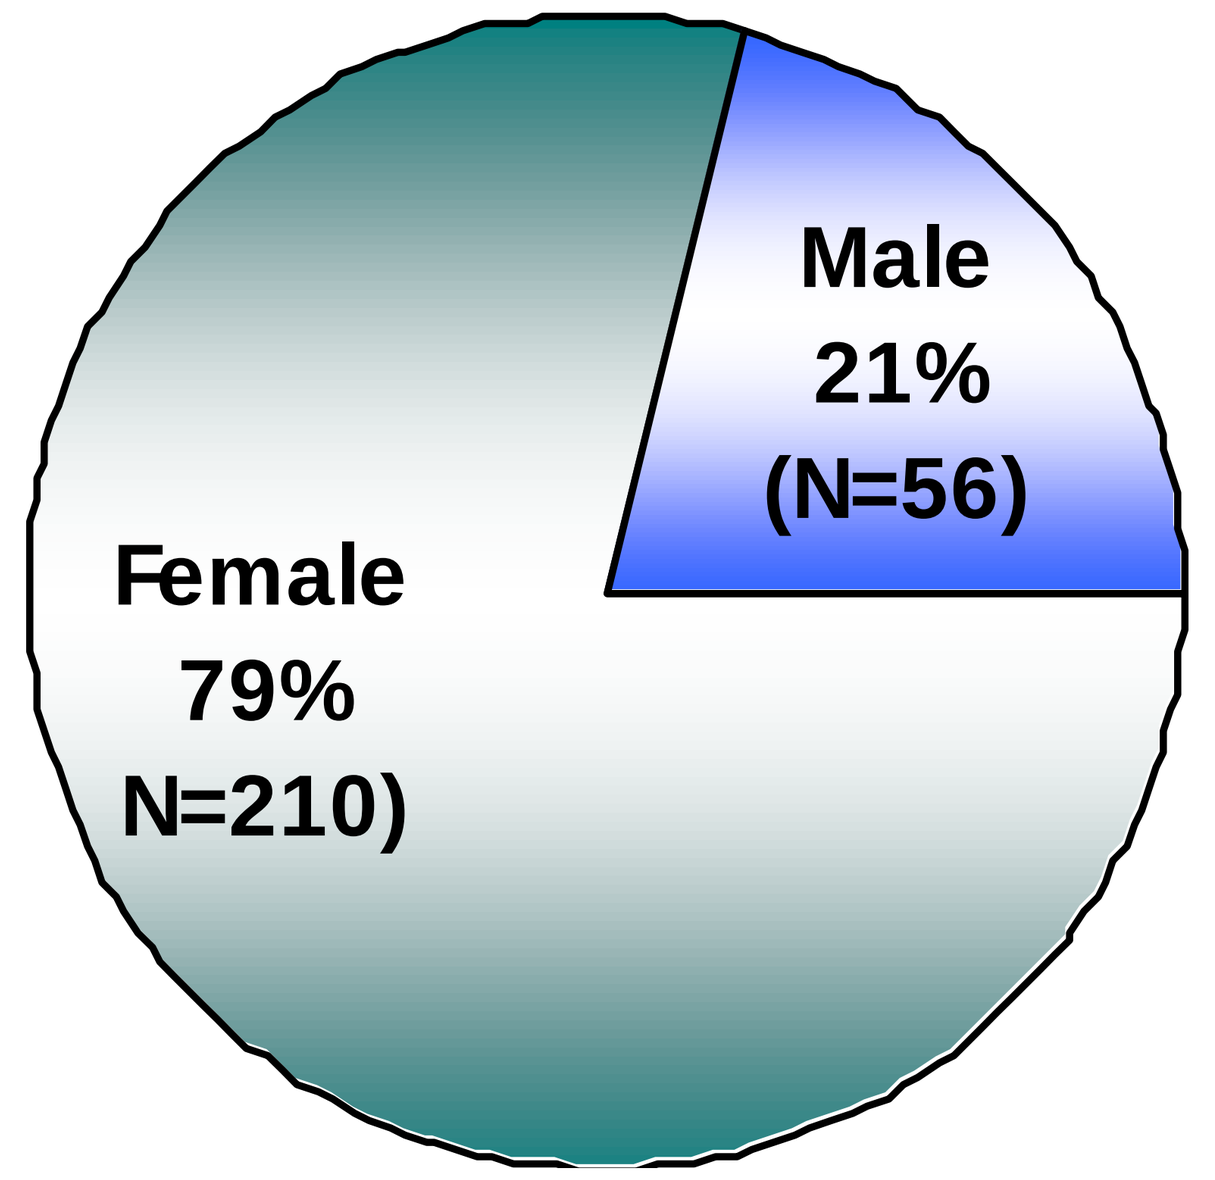
Male 21% (N=56)
Female 79% N=210)

Racial/ethnicity distribution of those receiving monthly depot leuprolide administrations during the index period (N=266)

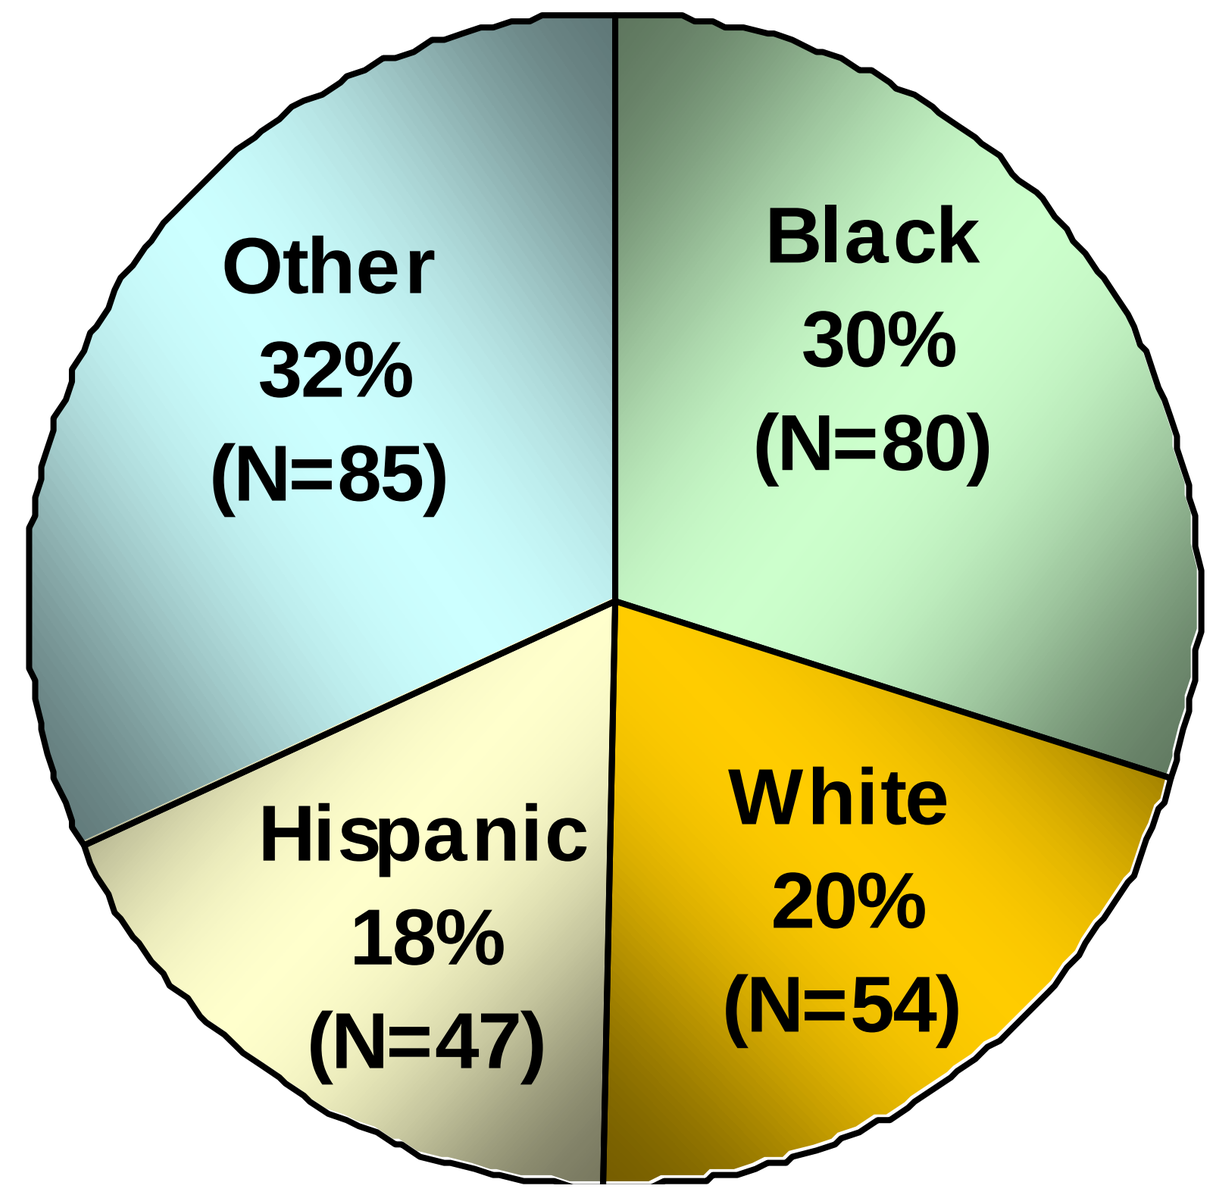
Other 32% (N=85)
Black 30% (N=80)
Hispanic 18% (N=47)
White 20% (N=54)

Average age distribution at 1st diagnosis among those receiving monthly depot leuprolide administrations during the index period (N=266)

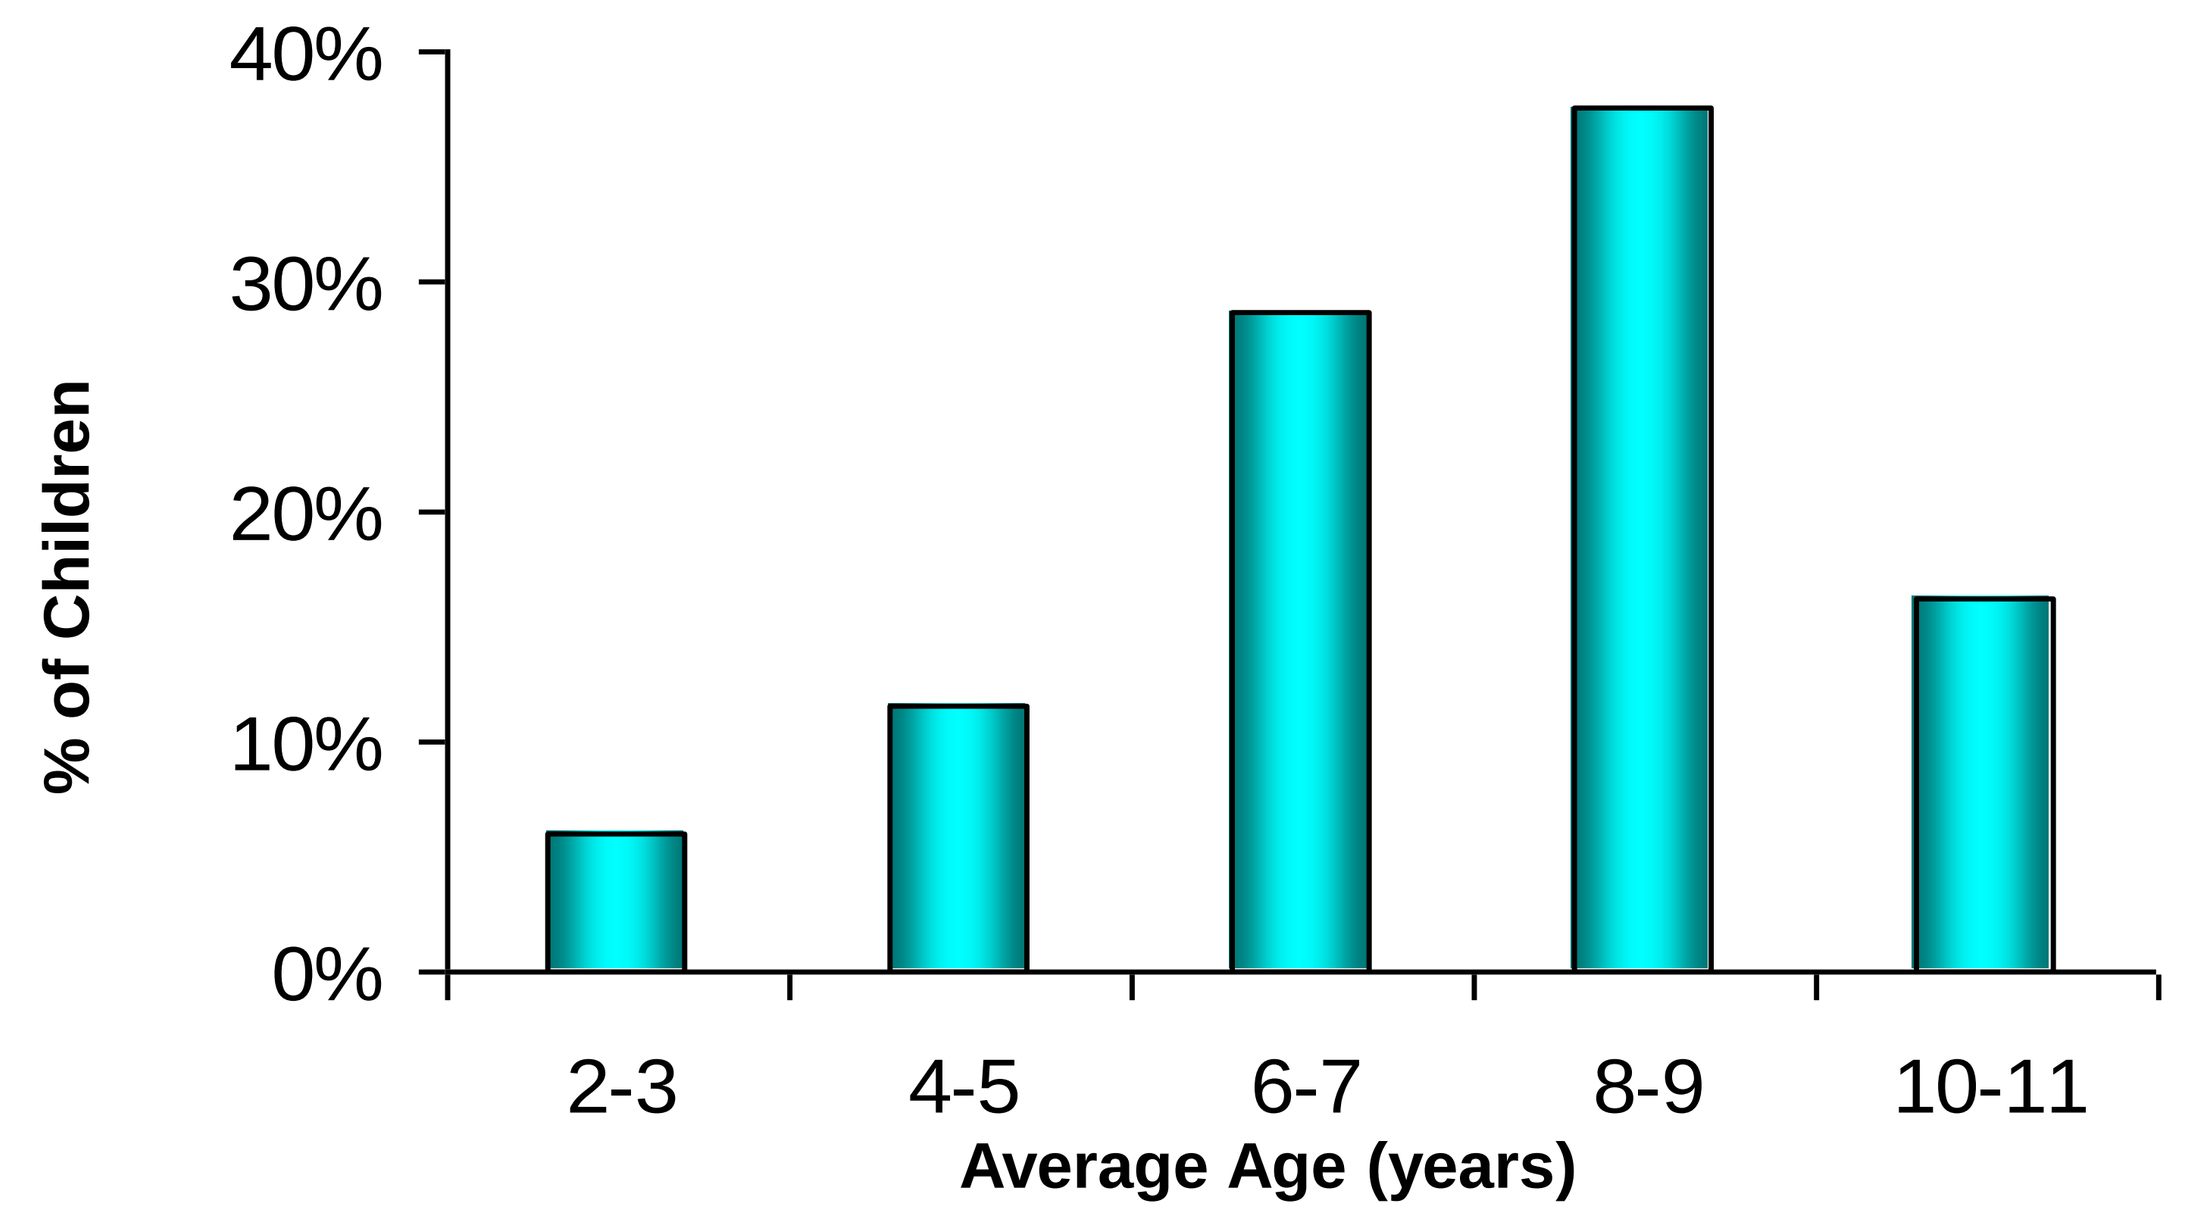
40%
30%
20%
10%
0%
% of Children
2-3
4-5
6-7
8-9
10-11
Average Age (years)

Average age at 1st CPP diagnosis was 7.4 (SD 2.2).

Among patients who received at least two administrations (N=243), average number of days between administrations

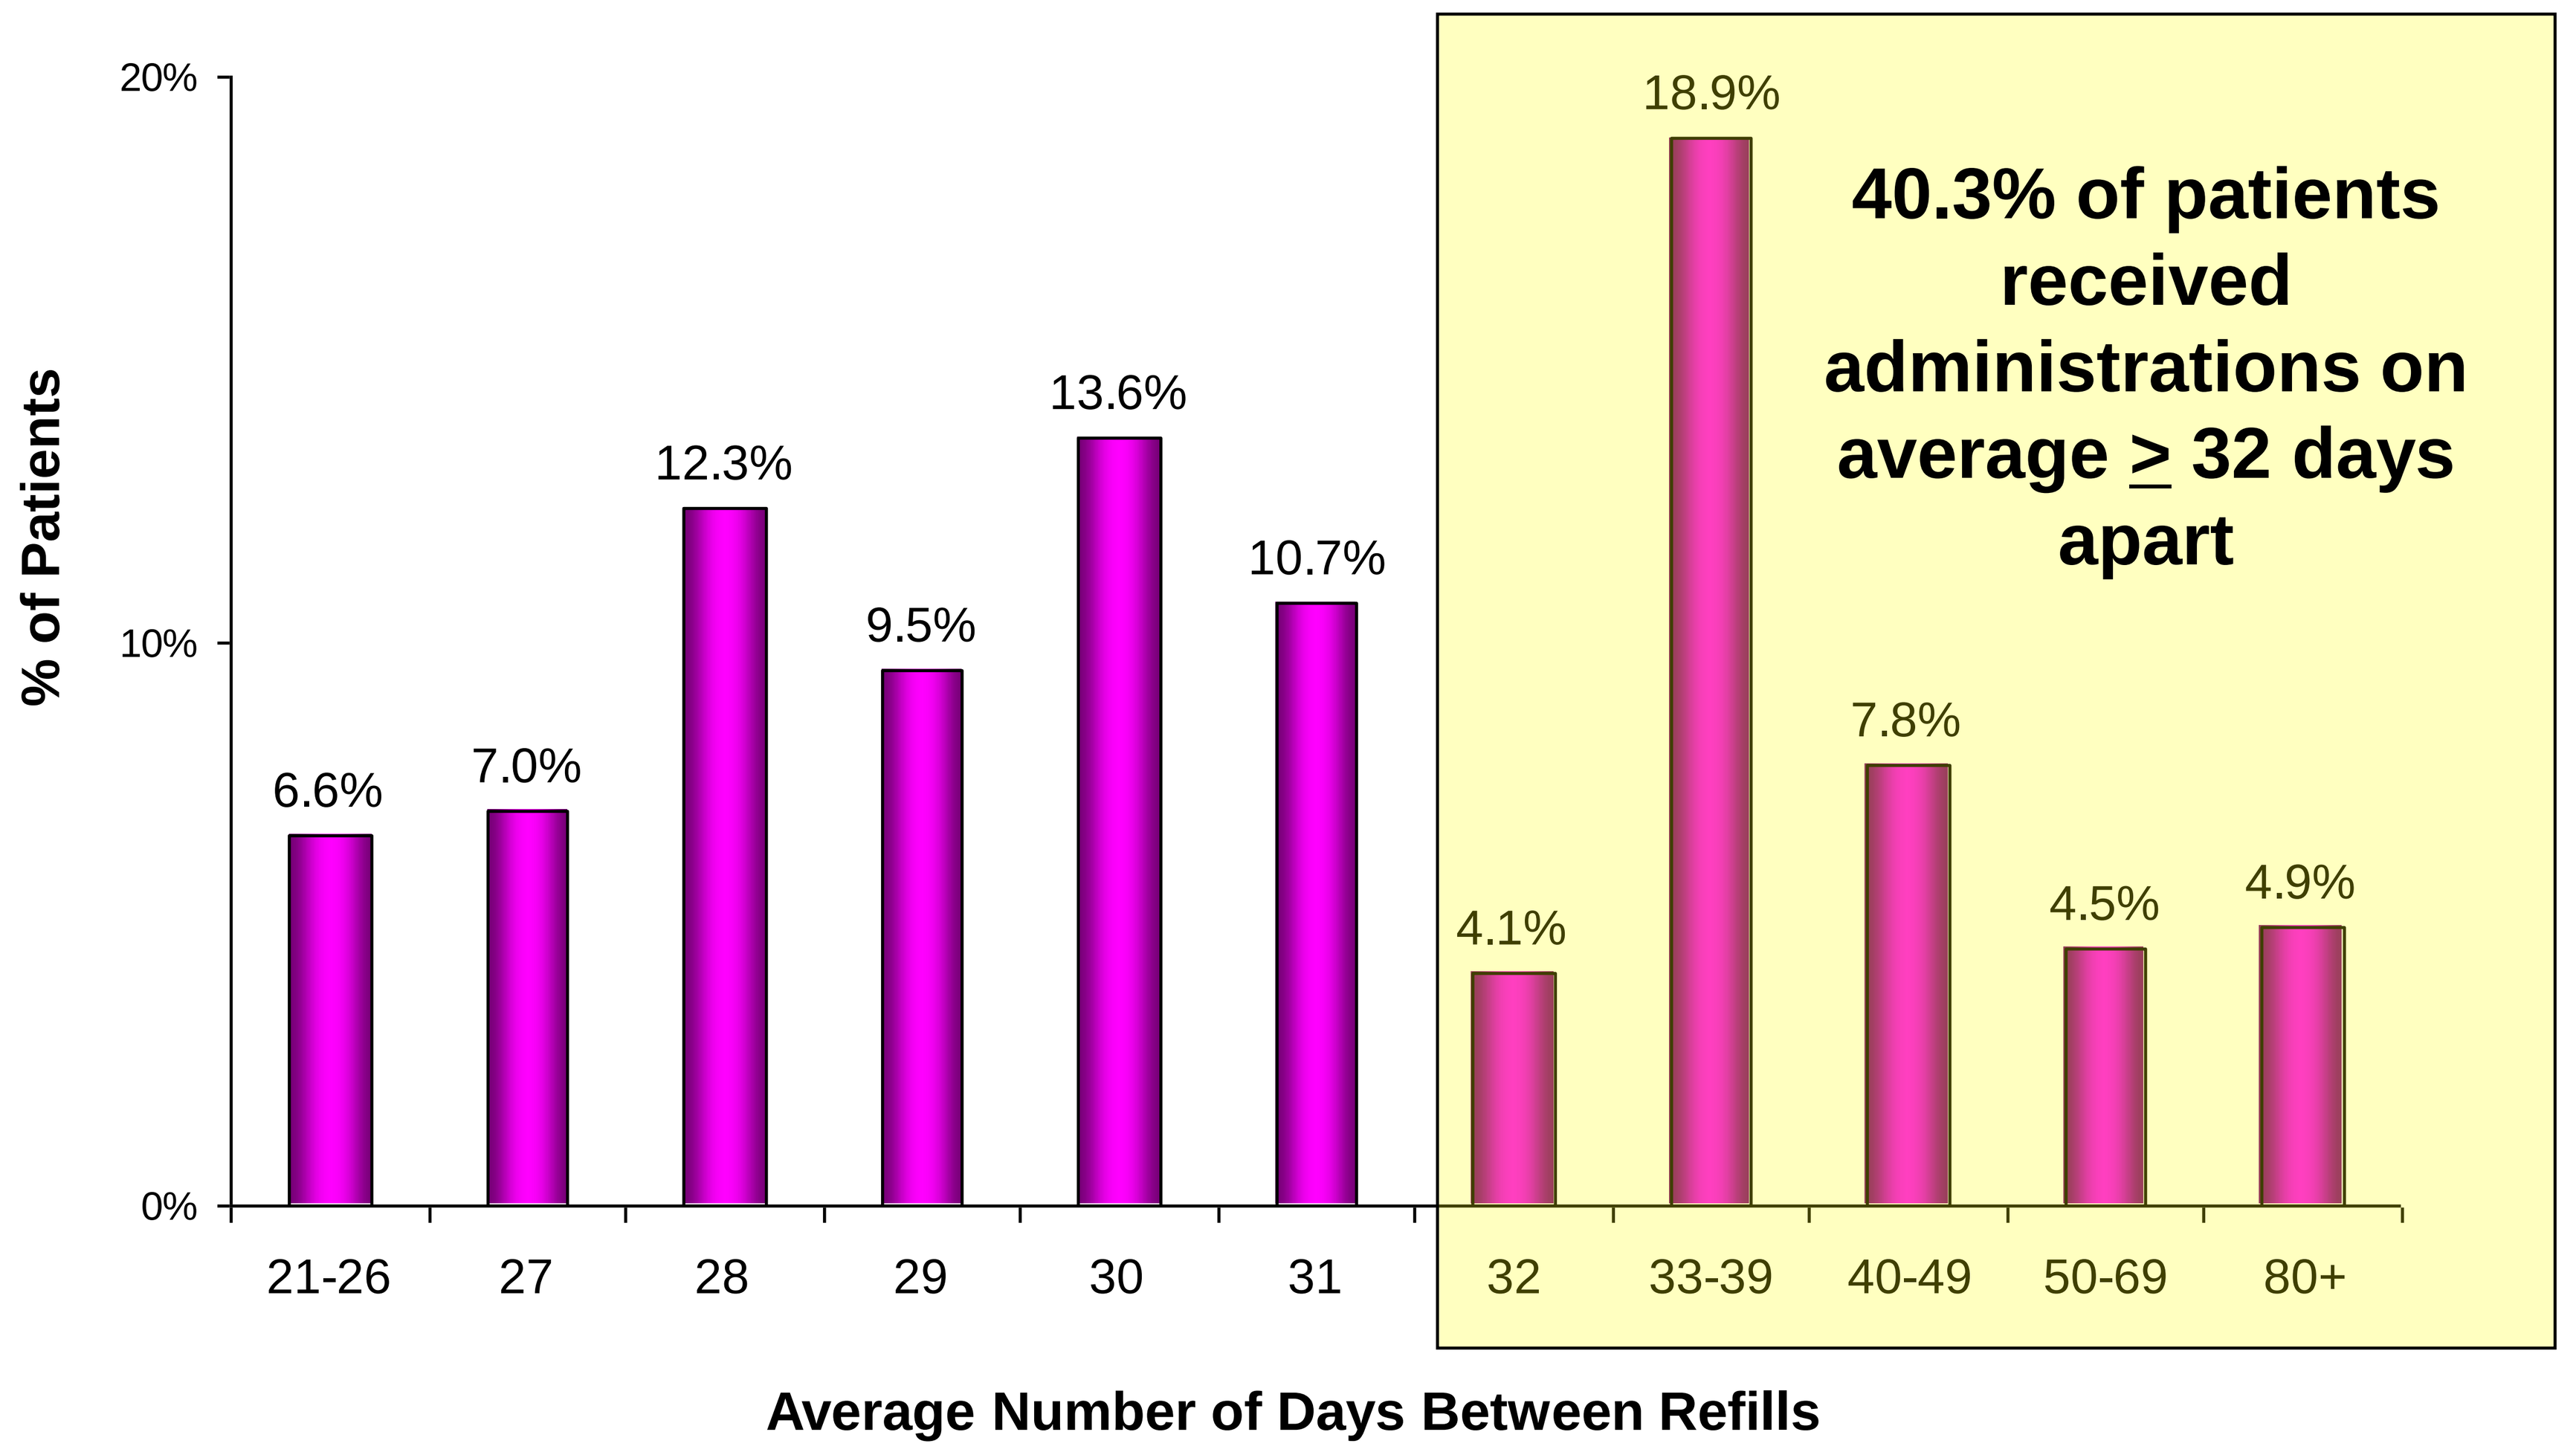
20%
10%
0%
% of Patients
6.6%
7.0%
12.3%
9.5%
13.6%
10.7%
4.1%
18.9%
7.8%
4.5%
4.9%
21-26
27
28
29
30
31
32
33-39
40-49
50-69
80+
40.3% of patients received administrations on average ≥ 32 days apart
Average Number of Days Between Refills

Among patients who received at least two administrations (N=243), the average number of days between administrations was 37 (SD 17.6), with a range of 21 to156 days.

CONCLUSIONS

Among Florida Medicaid-enrolled children diagnosed with CPP who received at least 2 administrations of monthly depot leuprolide, only (25.9%) received treatment within the 28-day recommended administration period.

Our findings suggest that the majority of children with CPP who receive monthly depot leuprolide may be inadequately controlled and are therefore at risk for the return of pubertal signs such as menses, breast development, testicular growth and long-term compromise of adult stature.

REFERENCES

1. Kaplan SL, Grumbach MM. Clinical review 14: Pathophysiology and treatment of sexual precocity. J Clin Endocrinol Metab 1990;71(4):785-9.
2. Deardorff J, Gonzales NA, Christopher FS, Roosa MW, Millsap RE. Early puberty and adolescent pregnancy: the influence of alcohol use. Pediatrics 2005;116(6):1451-6.
3. Offisioso A, Salerno M, Lettiero T, Michillo M, Pisatura L, Di Maio S. Adolescent girls with idiopathic central precocious puberty: typical character traits. J Pediatr Endocrinol Metab 2000;13(suppl 1):835-9.
4. Sonis WA, Comite F, Blue J, et al. Behavior problems and social competence in girls with true precocious puberty. J Pediatr 1985;106(1):156-60.
5. Michaud PA, Suris JC, Deppen A. Gender-related psychological and behavioural correlates of pubertal timing in a national sample of Swiss adolescents. Mol Cell Endocrinol 2006;25:254-5.
6. Carel JC, Lahlou N, Roger M, Chaussain JL. Precocious puberty and statural growth. Hum Reprod Update 2004;10(2):135-47.

FOR FURTHER INFORMATION

Please contact: Cheryl Hankin, PhD, BioMedEcon, LLC.; P.O. Box 129, Moss Beach, CA 94038; 650-563-9475; cherylhankin@biomedecon.com; www.biomedecon.com

DISCLOSURES

Authors of this presentation have the following to disclose concerning possible financial or personal relationships with commercial entities that may have a direct or indirect interest in the subject matter of this presentation:
Cheryl Hankin, Amy Bronstone, and Zhaohui Wang are Consultants to Indevus Corporation
Research supported by Indevus Corporation.

PRESENTED AT THE 42nd MIDYEAR CLINICAL MEETING OF THE AMERICAN SOCIETY OF HEALTH-SYSTEM PHARMACISTS, LAS VEGAS, DECEMBER 2-6, 2007.