Systematic Review and Cost-Effectiveness Analysis of Treatments for Moderate-to-Severe Psoriasis

CS Hankin PhD, BioMedEcon LLC, San Jose, CA; **SR Feldman MD PhD,** Wake Forest University School of Medicine, Winston-Salem, NC; **A Szczotka PharmD,** MedImpact Healthcare Systems Inc, San Diego, CA;
L Fish PharmD, Fallon Community Health Plan, Worcester, MA; **E Scharaga RPh,** Group Health Inc, New York, NY

BACKGROUND

Psoriasis is a skin disorder characterized by scaly plaques, itching, and redness. Of the 8 million American sufferers, 30% require phototherapy (ultraviolet B light or psoralen with ultraviolet A light), oral systemics (acitretin, cyclosporine, and methotrexate), and/or biologic treatments (which include alefacept, efalizumab, etanercept, and infliximab).[1,2]

Since 2003, the number of FDA-approved biologic treatments for psoriasis has increased three-fold. As burgeoning demand for these newer and more expensive drugs contends with limited health systems resources, US healthcare decision makers will need to consider ways in which to compare efficacy and costs across the range of treatments.

OBJECTIVE AND PERSPECTIVE

The Psoriasis Area Severity Index (PASI) is the most frequently cited measure of efficacy, and is accepted by the FDA as a primary outcome.[3,4] PASI may be reported in terms of average percentage change (PASI%) from baseline to endpoint. Clinically meaningful outcomes are frequently reported as at least 50% or 75% improvement from baseline ("PASI 50" or "PASI 75").

We compared the cost-effectiveness of phototherapy, oral systemic, and biologic outpatient treatments for moderate-to-severe psoriasis in terms of PASI% improvement. Specifically, we asked: From the perspective of the US healthcare system (i.e., physicians, pharmacy and therapeutics committee members, medical directors, clinical pharmacists, and other healthcare professionals), what are the comparative annual costs to achieve PASI improvement (1%, 50%, and 75%) among these targeted psoriasis treatments?

METHODS

- Expert Panel in dermatology, health services research and managed care conducted a systematic literature review (Medline search 1966 – August 2004) of treatment efficacy for moderate-to-severe psoriasis in terms of average percentage PASI improvement.
- Inclusion / exclusion criteria were designed to minimize the introduction of bias (see Table 1).
- Priority was given to randomized, double-blind, placebo-controlled trials.
- Treatments included acitretin, alefacept, cyclosporine, efalizumab, etanercept, infliximab, methotrexate, narrowband ultraviolet B (NBUVB), broadband ultraviolet B (BBUVB), psoralen with ultraviolet A (PUVA), BBUVB combined with acitretin, and PUVA combined with acitretin.
- Applied January 2004 US Average Wholesale Drug Prices (AWP) and 2004 Medicare Reimbursement Rates (MRR) to calclulate cost-effectiveness (CE) as:

CE = [(Total Costs of Drug at AWP or UV at MRR) + (Costs of Administration at MRR) + (Costs of Screening at MRR) + (Risk Adjusted Costs of Adverse Events at MRR)] / (Average PASI% per Systematic Literature Review)

Table 1: SYSTEMATIC LITERATURE REVIEW - STUDY EXCLUSION CRITERIA

- PASI % improvement from baseline not reported or unavailable for calculation
- Number of Ss receiving active treatment <10
- Treatment duration <6 or > 14 wks
- Diagnosis other than moderate-to-severe psoriasis or PASI < 8
- Sample does not consist predominantly of plaque psoriasis
- Mean dose unspecified
- For monotherapies, concomitant systemic or phototherapy psoriasis medication/treatment was received in addition to targeted treatment
- Maintenance following stabilization (rather than de novo) treatment
- Dose exceeds current recommendation
- Duration of study or time at endpoint evaluation unspecified
- Study is duplication and previously reviewed by us
- Poor Study Quality (did not apply Intent-to-Treat)
 - Disposition of dropouts unclear or dropouts excluded from analysis
- Biased sample selection (e.g., Ss excluded from participation if poor response to treatment previously)
- Study Design
 - Retrospective review or Case Study

RESULTS

Among 3,886 articles documented in MEDLINE (1966 through August 2004), we identified 16 articles (for which there were 18 outcomes) to include in our calculation of PASI% (Table 2). Shown below are annualized costs of: drug or UV treatment (Table 2), risk-adjusted costs of adverse events (Table 3), costs of administration of biologics (Table 4), and costs of screening (Table 5). Also shown are annualized total costs of treatment (Figure 2), average PASI % (Figure 3), annualized costs to achieve 1% (Figure 4) and 75% (Figure 5) PASI improvement.

The costs to achieve 1% PASI improvement were $31 for methotrexate 7.5 mg, and $33 methotrexate 15 mg. The cost to achieve 1% PASI improvement for PUVA ranged from $41 to $63, BB UVB from $56 to $100, and NB UVB $64. Combined treatment of BB UVB with acitretin 25 mg provided 1% PASI improvement at a cost of $55, and PUVA with acitretin 20 mg $76. The cost to achieve 1% PASI improvement for cyclosporine was $134 and $141 for 3 mg/kg/day and 1.5 mg/kg/day respectively. Acitretin 50mg/day provided 1% PASI improvement at a cost of $216, infliximab 5 mg/kg $319, etanercept 50 mg $330, efalizumab 1 mg/kg $346, and alefacept 15 mg IM $602.

Table 2: PASI % AND TOTAL ANNUAL DRUG OR UV TREATMENT COSTS

		ANNUALIZED TREATMENT DOSE AND DURATION		Total Annual $Rx or $UV Costs
Treatment	PASI %	Trial Dose and Duration	Assumed Maintenance Dose & Duration	
BBUVB	80.1%[5]	3x/wk, 12 wks	2x/wk for 40 wks	$4,807[a]
BBUVB	47.0%[6]	3x/wk, 6 wks	2x/wk for 46 wks	$4,558[a]
BBUVB	83.7%[7]	3x/wk, 7 wks	2x/wk for 45 wks	$4,600[a]
NBUVB	73.0%[6]	3x/wk, 6 wks	2x/wk for 46 wks	$4,558[a]
PUVA	92.9%[8]	4x/wk, 6 wks; 2x/wk, 2wks	1x/wk for 44 wks	$3,737[b]
PUVA	61.1%[9]	3x/wk,10 wks	1x/wk for 42 wks	$3,737[b]
acitretin	60.4%[10]	48.2 mg/day	constant dose	$12,359[c]
cyclosporine	52.0%[11]	3 mg/kg/day	constant dose	$5,019[d]
cyclosporine	33.4%[12]	1.5 mg/kg/day	constant dose	$2,789[e]
methotrexate	58.4%[13]	7.5 mg/wk	constant dose	$595[f]
methotrexate	71.6%[13]	15 mg/wk	constant dose	$1,190[f]
alefacept	45.0%[14]	15 mg/wk	2 12-week courses	$23,88 0[g]
efalizumab	52.0%[15]	1 mg/kg/wk	constant dose	$17,836[h]
etanercept	64.2%[16]	50 mg 2x/wk, 12 wks	25 mg/wk 2x/wk, 40 wks	$21,052[i]
infliximab	82.8%[17]	5 mg/kg wks 0, 2, 6	6 administrations	$24,898[j]
UVB + acitretin	80.7%[18]	UVB 5 – 7/wk + 25 mg acitretin/d, for 12 wks	UVB 0.5 x/wk, acitretin 12.5mg/d, 40 wks	$3,792[f]
PUVA + acitretin m	97.3%[19]	acitretin 40mg/d, 2 wks; 20mg/dy+PUVA 3wk,8 wks	PUVA 0.5x/wk, acitretin 12.5 mg/d, 42 wks	$6,773[b,h]

[a] CPT 96910 ($41.44) ; [b] CPT 96912 ($51.90); [c] 2-25mg tabs; [d] 225 mg/d of 2-100mg tabs +1-25mg tab; [e] 125 mg /d, of 1-100mg tab + 1-25mg tab; [f] single tab; g single dose; [h] 75mg /wk of 1 vial with 50mg waste; [i] ea vial of 25 mg; [j] 375mg/admin of 4 vials w/ 25mg waste; [k] 1-25mg, 1-10mg, ½-10mg tabs; [l] commercial tanning beds.

Table 3: ANNUALIZED RISK-ADJUSTED COSTS OF ADVERSE EVENTS

	Risk of Adverse Event	Costs of Adverse Events	Annualized Risk-Adjusted Costs of Adverse Events
UVB	NMSC: 2% annual incidence. [20]	$875[a] /year[28]	($875 * 2%) = **$18**
PUVA	NMSC: 5-year risk is 1.2%. [21, 22]	$875[a] /year[28]	($875 * 1.2%) = **$11**
acitretin	Hyperlipidemia: 8.25% of patients will require lipid-lowering agents. [23, 24, 25]	20 mg atorvastatin calcium: $2.76/day (AWP) [29]	($2.76 * 8.25%) * 365 days = **$83**
cyclosporine	Hypertension: 18% of patients will require calcium channel antagonists. [26]	300 mg long-acting diltiazem HCL: $2.10/day (AWP) [29]	($2.10 *17.75%) * 365 days = **$138**
methotrexate	Unknown at doses below 20 mg/wk	-	-
alefacept	unknown	-	-
efalizumab	unknown	-	-
etanercept	unknown	-	-
infliximab	unknown	-	-
UVB + acitretin	Hyperlipidemia: 4.13% of patients will require lipid-lowering agents. [23, 24, 25] NMSC: 5-year risk is 1%.[20] Acitretin reduced risk 70%[27]	20 mg atorvastatin calcium: $2.76/day (AWP) [29]+ $875[a] /year[28]	[($2.76 * 4.13% * 365 days] + ($875 * 2% * 70%) = **$54**
PUVA + acitretin	Hyperlipidemia: 4.13% of patients will require lipid-lowering agents. [23, 24, 25] NMSC: 5-year risk is 2%.[21, 22] Acitretin reduced risk 70%[27]	20 mg atorvastatin calcium: $2.76/day (AWP) [29]+ $875[a] /year[28]	[($2.76 * 4.13% * 365 days] + ($875 * 1.2% * 70%) = **$49**

Table 4: ANNUALIZED COSTS OF TREATMENT ADMINISTRATION

Biologic Drug	Mode of administration	Can drug be patient-administered?	Number of clinician admin/ yr	Evaluation and Management	2004 Medicare Reimbursement Rate	Total Annualized Costs
alefacept	IM bolus	No	24	90782	$33.60	$806
efalizumab	SC	Yes	0	-	-	0
etanercept	SC	Yes	0	-	-	0
infliximab	IV infusion 2 hours	No	9	90780, 90781	$157.99	$1,422

Table 5: ANNUALIZED COSTS OF SCREENING (per Product Labeling)

	Complete Physical	Brief Exam	Liver Fx	Lipid Panel	Serum Creat	CD4 T-Lymp	Electrolytes	Mg	BUN	Uric Acid	CBC	RBC auto	TB Skin	Renal Fx-am	Chest X-Ray	Liver Biopsy	
2004 Medicare Rates ($)	$106	$8	$15	$19	$10	$89	$13	$13	$7	$9	$15	$8	$10	$16	$33	$684 $200	Total Annual Cost
BBUVB	1																$106
NB UVB	1																$106
PUVA	1																$106
acitretin	1		15	15													$618
cyclosporine	1	16	13	16	17		16	16	16	13	16						$1,794
methotrexate	1		13								13			13	1	0.5	$1,188
alefacept	1					26											$2,412
efalizumab	1											7					$163
etanercept	1																$106
Infliximab	1												1				$116
UVB + acitretin	1		15	15													$618
PUVA + acitretin	1		15	15													$618

Figure 1:
Annualized Total Costs (USD)
($ Rx/Tx + $ Admin + $ Screen + $ AE)

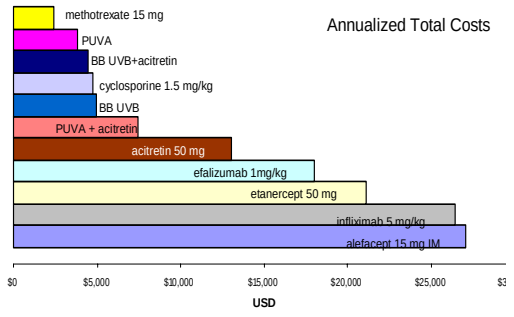
Annualized Total Costs
methotrexate 15 mg
PUVA
BB UVB+acitretin
cyclosporine 1.5 mg/kg
BB UVB
PUVA + acitretin
acitretin 50 mg
efalizumab 1mg/kg
etanercept 50 mg
infliximab 5 mg/kg
alefacept 15 mg IM
$0
$5,000
$10,000
$15,000
$20,000
$25,000
USD

Figure 2:
Reported Average PASI % Improvement from Baseline (% Efficacy)

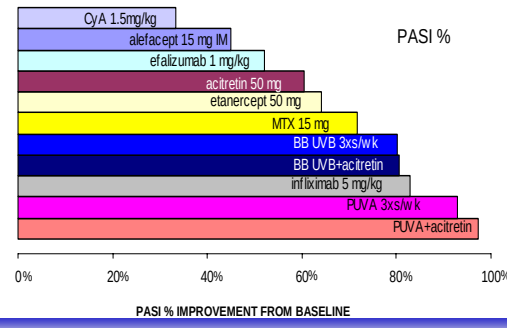
PASI %
CyA 1.5mg/kg
alefacept 15 mg IM
efalizumab 1 mg/kg
acitretin 50 mg
etanercept 50 mg
MTX 15 mg
BB UVB 3xs/wk
BB UVB+acitretin
infliximab 5 mg/kg
PUVA 3xs/wk
PUVA+acitretin
0%
20%
40%
60%
80%
100%
PASI % IMPROVEMENT FROM BASELINE

Figure 3:
Annualized Costs to Achieve 1% PASI Improvement (USD)

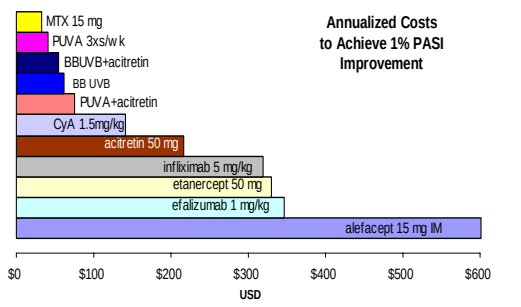
Annualized Costs to Achieve 1% PASI Improvement
MTX 15 mg
PUVA 3xs/wk
BBUVB+acitretin
BB UVB
PUVA+acitretin
CyA 1.5mg/kg
acitretin 50 mg
infliximab 5 mg/kg
etanercept 50 mg
efalizumab 1 mg/kg
alefacept 15 mg IM
$0
$100
$200
$300
$400
$500
$600
USD

Figure 4:
Annualized Costs to Achieve 75% PASI Improvement (USD)

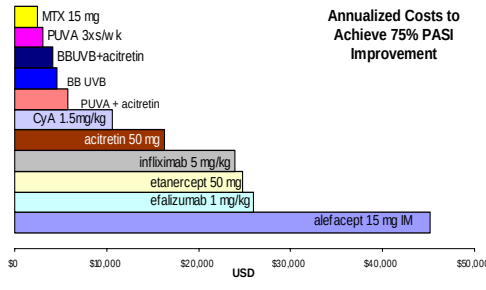
Annualized Costs to Achieve 75% PASI Improvement
MTX 15 mg
PUVA 3xs/wk
BBUVB+acitretin
BB UVB
PUVA + acitretin
CyA 1.5mg/kg
acitretin 50 mg
infliximab 5 mg/kg
etanercept 50 mg
efalizumab 1 mg/kg
alefacept 15 mg IM
$0
$10,000
$20,000
$30,000
$40,000
$50,000
USD

CONCLUSIONS

Oral systemics, UV therapy, and combined UV therapy with acitretin appear to be the most cost-effective treatments for moderate-to-severe psoriasis. Findings suggest that the most costly medications were not necessarily the most efficacious.

REFERENCES

1. Krueger. *Arch Dermatol*. 2001;137:280-284. **2.** Koo. *Dermatol Clin*. 1996;14:485-96. **3.** Fredriksson. *Dermatologica*. 1977; 157;238-244. **4.** Weisman. *J Dermatol Treat*. 2003;14:158-165. **5.** Ramsay. *Dermatology*. 2000;200(1):17-24. **6.** Walters. *J Am Acad Dermatol*. 1999; 40:893-900. **7.** Woo. *Br J Dermatol*. 2003 Jul;149(1):146-50. **8.** Caca-Biljanovska. *Croat Med J*. 2002 Dec;43(6):707-12. **9.** Torras. *J Dermatolog Treat*. 2004 Apr;15(2):98-103. **10.** Gollnick. *Eur J Dermatol*. 1993;3:442-6. **11.** Ellis. *N Eng J Med*. 1991;324:277-84. **12.** Reitamo. *Br J Dermatol*. 2001 Sep;145(3):438-45. **13.** Chladek. *Br J Clin Pharmacol.* 2002 Aug;54(2):147-56. **14.** Lebwohl. *Arch Dermatol*. 2003;139:719-727. **15.** Gordon. *JAMA*. 2003;290:3073-3080. **16.** Leonardi. *N Engl J Med*. 2003 Nov 20;349(21):2014-22. **17.** Chaudhari. *Lancet*. 2001;357:1842-47. **18.** Carlin. *Arch Dermatol*. 2003 Apr;139(4):520-4. **19.** Lauharanta. *Br J Dermatol*. 1989 Jul;121(1):107-12. **20**. Pasker-de Jong. *Arch Dermatol*. 1999;135:834-40. **21**. Morison. In: Weinstein GD, Gottleib AB, eds. New York: Marcel Dekker, Inc, 2003:91-114. **22**. Miller. *J Am Acad Dermatol*. 1994 May;30(5 Pt 1):774-8. **23**. Soriatane [package insert]. Palo Alto, CA: Connetics Inc; 2004. **24**. McClure. *Drug Safety*. 2002;25:913-927. **25**. Katz. *J Am Acad Dermatol*. 1999;41:S7-12. **26.** Lebwohl. *J Am Acad Dermatol*. 1998;39:464-75. **27.** Nijsten. *J Am Acad Dermatol*. 2003;49:644-50. **28.** Housman. *J Am Acad Dermatol*. 2003;48:425-9. **29**. *Red Book*. 2004 Edition. Montvale, NJ: Thompson Micromedex; 2004.

Disclosures: Dr. Hankin is a consultant for Connetics Corporation. **Funding Source:** This study was funded by Connetics Corporation.